Abstract #238

Azenosertib in Combination With Bevacizumab as Maintenance Therapy in Patients With Advanced Ovarian Cancer: Part 2 of a Phase 1b study (MUIR/Zn-c3-002)

Joyce F. Liu[1], Tarek Meniawy[2], Gary Richardson[3], Siqing Fu[4], Premal H. Thaker[5], Stephanie V. Blank[6], Cara Mathews[7], Angeles Alvarez Secord[8], Bradley Corr[9], Ashwin Swami[10], Catherine Lee[10], Christine Burnett[10], Dongliang Zhuang[10], Nandini Molden[10], Sumitra Ananda[11]

[1]Dana-Farber Cancer Institute, Boston, MA; [2]Linear Cancer Research, Perth, Australia; [3]Cabrini Hospital, Malvern, Australia; [4]MD Anderson Cancer Center, Houston, TX; [5]Washington University St. Louis and Siteman Cancer Center, St. Louis, MO; [6]Icahn School of Medicine at Mount Sinai, New York, NY; [7]Women & Infants Hospital, Legorreta Cancer Center, Alpert Medical School of Brown University, Providence, RI; [8]Duke Cancer Institute, Durham, NC; [9]University of Colorado, Aurora, CO; [10]Zentalis Pharmaceuticals, Inc., San Diego, CA; [11]Epworth Healthcare, Melbourne, Australia

INTRODUCTION/BACKGROUND

- Surgical debulking followed by adjuvant carboplatin-paclitaxel chemotherapy remains the standard approach for patients with Stage III/IV ovarian cancer (OC)
- Most patients ultimately experience relapsed disease within the first 5 years due to the advanced stage at diagnosis and face an unfavorable prognosis[1]; effective novel targeted therapies are needed to address this substantial unmet need
- WEE1 tyrosine kinase is a key regulator of multiple checkpoints that prevents progression through the cell cycle, allowing time for DNA repair before cell division[2,3]
- Inhibition of WEE1 tyrosine kinase results in unscheduled mitosis and eventually cancer cell death[3,4]
- Azenosertib (**Figure 1**), a potential best-in-class, small molecule, highly selective oral WEE1 inhibitor demonstrated clinically meaningful activity in Cyclin E1 positive platinum-resistant, high-grade serous OC (HGSOC) in the DENALI Part 1b clinical trial (NCT05128825)[5]
- Maintenance therapy with a poly(ADP-ribose) polymerase inhibitor (PARPi) following 6 cycles of platinum-based chemotherapy is a standard of care for many patients with advanced HGSOC[6], and has demonstrated significantly prolonged progression-free survival in patients with platinum-sensitive, recurrent OC[7]
 - Nearly half of patients relapse while on first-line maintenance with a PARPi[8], and outcomes are poor with a time from first to second treatment of 6.1 months vs 11.4 months in patients who progressed after first-line maintenance[6], representing a significant therapeutic challenge in this patient population
- Bevacizumab is an anti-vascular endothelial growth factor (VEGF) monoclonal antibody approved for the maintenance of advanced OC following platinum-based chemotherapy[9,10]
- Distinct mechanisms of action and limited overlapping toxicities provide a strong rationale for adding azenosertib to bevacizumab as maintenance therapy in HGSOC
- Combining azenosertib with anti-VEGF antibody was well tolerated and showed enhanced activity in PSOC preclinical models[11]
- This combination regimen is currently being evaluated in the Zn-c3-002 study (MUIR; NCT04516447)

Figure 1. Mechanism of action of azenosertib

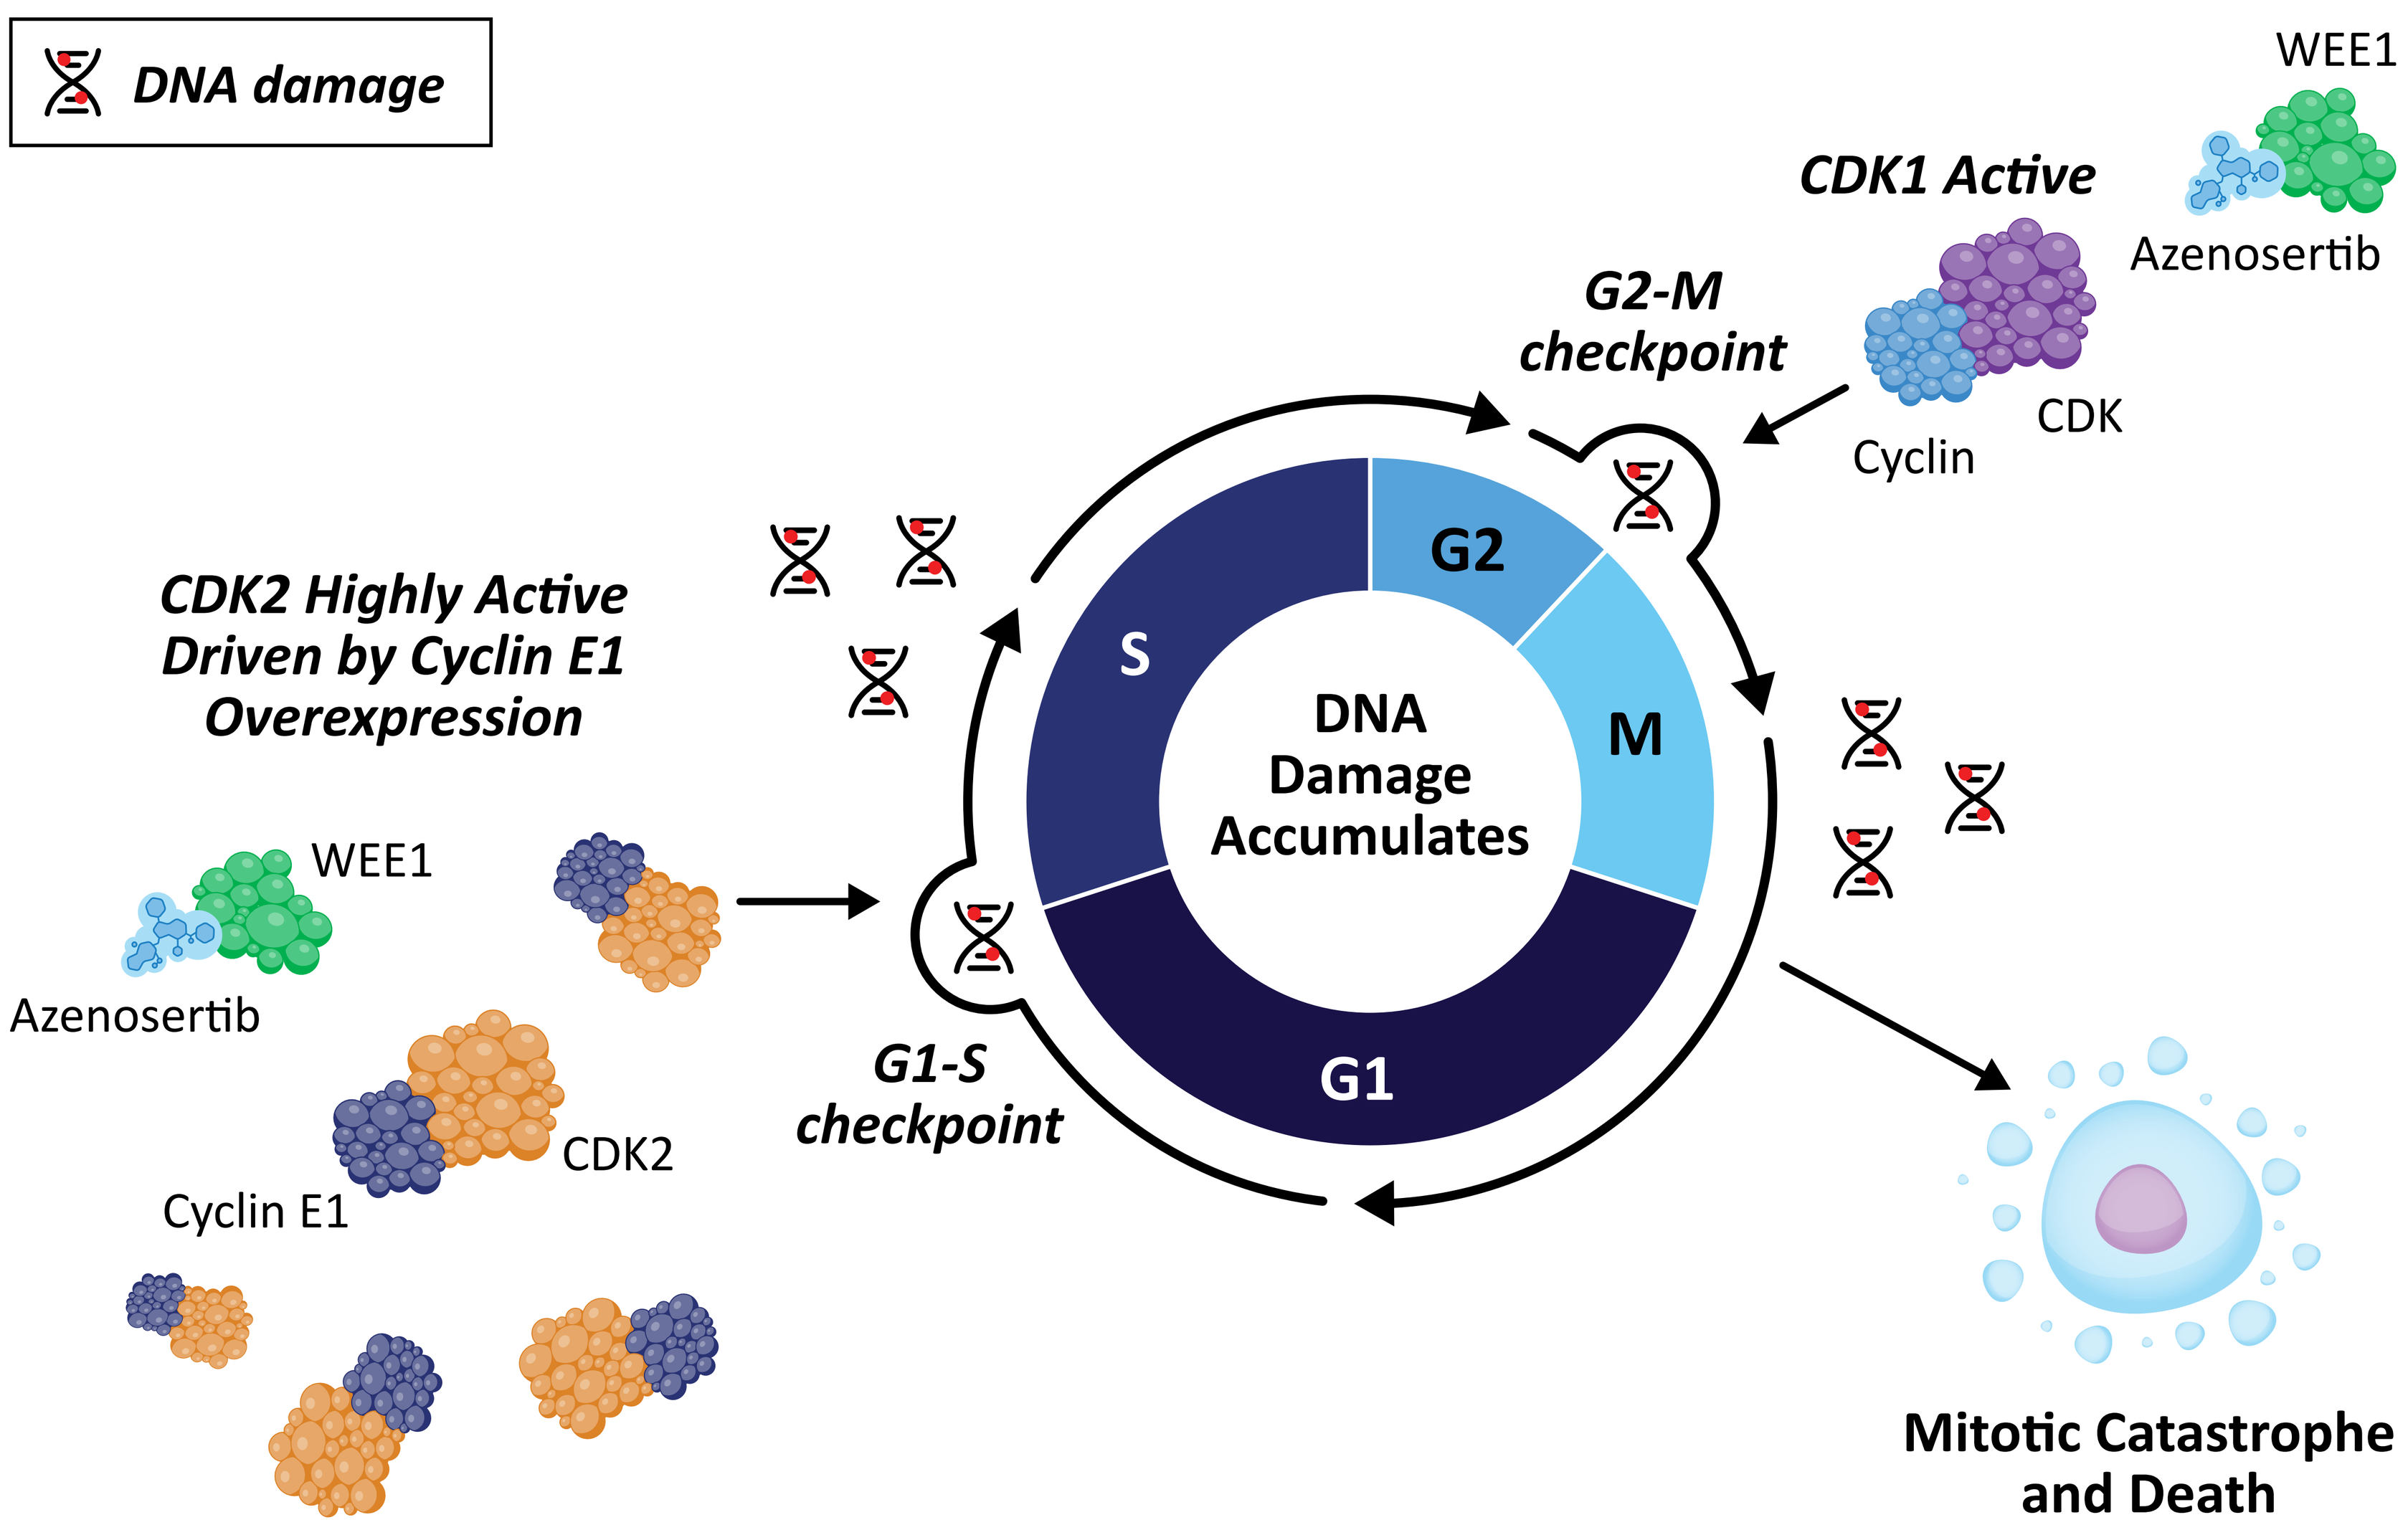

DNA damage
WEE1
Azenosertib
CDK1 Active
G2-M checkpoint
Cyclin
CDK
CDK2 Highly Active Driven by Cyclin E1 Overexpression
WEE1
Azenosertib
CDK2
Cyclin E1
G1-S checkpoint
S
G2
M
G1
DNA Damage Accumulates
Mitotic Catastrophe and Death

METHODOLOGY

Figure 2. Study Design of MUIR – Part 2

DOSE ESCALATION (up to 12 patients)

Key Eligibility Criteria
- Age ≥18 years
- ECOG PS 0-1
- Histologically/cytologically confirmed FIGO Stage III/IV ovarian, peritoneal, or fallopian tube cancer
- 6 cycles of platinum-based chemotherapy
 - **1L:**
 - No germline/somatic *BRCA* mutation
 - CR, PR, or SD
 - **2L:**
 - Prior PARPi for patients with *BRCA* mutation
 - PD >183 days after last dose in 1L, and CR, PR, or SD to 2L

DL 1 (n=3-6)
Azenosertib **300 mg** PO QD 5:2 in 21-day cycles[a]
+
Bevacizumab 15 mg/kg IV on Day 1 of each 21-day cycle[b]

DLTs in ≤1 of 6 patients

DL 2 (n=3-6)
Azenosertib **350 mg** PO QD 5:2 in 21-day cycles[a]
+
Bevacizumab 15 mg/kg IV on Day 1 of each 21-day cycle[b]

Recommended dose for dose expansion

DOSE EXPANSION (up to 40 patients)

Key Eligibility Criteria
- Age ≥18 years
- ECOG PS 0-1
- Histologically/cytologically confirmed FIGO Stage III/IV ovarian, peritoneal, or fallopian tube cancer
- Progression while on a PARPi in 1L maintenance
- PD >183 days post last dose of 1L chemotherapy, and CR, PR or SD after ≥4 cycles of platinum-based chemotherapy in 2L

Treatment:
Azenosertib at recommended dose from Dose Escalation
+
Bevacizumab

Primary objectives:
- Safety/tolerability
- Recommended dose for dose expansion (dose escalation only)

Secondary objectives:
- Clinical activity
- PK of azenosertib and metabolites

Duration of Patient Participation

Screening Up to 28 days → **21-day treatment cycles** → **EOT visit** Within 7 days after last dose → **Safety follow-up visit** 30 (± 7 days after last dose) → **Disease assessment follow-up visits** Every 6 weeks (± 7 days) in dose expansion until radiological confirmation of PD, or other protocol-specified reasons

[a]Up to 24 months. [b]Up to 15 months.

- Zn-c3-002 (NCT04516447; MUIR) is an open-label, 2-part, Phase 1b study, evaluating preliminary efficacy, safety, and pharmacokinetics of azenosertib combination regimens in patients with OC
- In Part 1, patients receive azenosertib with investigator's choice of chemotherapy (pegylated liposomal doxorubicin, carboplatin, paclitaxel, or gemcitabine)
- In **Part 2,** patients receive azenosertib in combination with bevacizumab (**Figure 2**)
- **Primary objectives**:
 - Safety/tolerability assessed by:
 - Incidence and severity of adverse events
 - Incidence of dose interruptions, reductions and discontinuations due to treatment-related adverse events
 - Recommended dose for dose expansion (dose escalation only)
- **Secondary objectives**:
 - Clinical activity assessed by:
 - ORR, CBR, and DOR[a]
 - PFS
 - Time to progression by CA-125 assessment
 - PK of azenosertib and metabolites
- **Exploratory objectives**:
 - Association between sensitivity to azenosertib and molecular characteristics of tumor tissue and plasma including:
 - Cyclin E1 expression
 - Gene mutations, insertions, deletions, copy number variations, and structural variants
 - Gene expression signatures

[a]For patients who enter the study with measurable disease.

Trial Status

Dose escalation is ongoing; dose expansion will enroll in Europe, US, and Australia

References

1. Kurnit KC, et al. *Obstet Gynecol.* 2021;137:108-121.
2. Ghelli Luserna di Rorà A, et al. *J Hematol Oncol*. 2020;13(1):126.
3. Esposito F, et al. *Int J Mol Sci*. 2021;22(19):10689.
4. Vriend LE, et al. *Biochim Biophys Acta*. 2013;1836(2):227-235.
5. Simpkins F, et al. Presented at the Society of Gynecologic Oncology Annual Meeting, March 14-17, 2025, Seattle, WA.
6. Harter P, et al. *Ann Oncol*. 2025;36(2):185-196.
7. Mirza MR, et al. *N Engl J Med*. 2016;375(22):2154-2164.
8. Loverro M, et al. *ESMO Open*. 2025;10(2):104119.
9. AVASTIN Prescribing Information. Genentech, Inc. 2022.
10. Perren TJ, et al. *N Engl J Med*. 2011;365:2484-2496.
11. Abed M, et al. Presented at the AACR-NCI-EORTC International Conference on Molecular Targets and Cancer Therapeutics, October 22-26, 2025, Boston, MA.

Acknowledgments

This study is sponsored by Zentalis Pharmaceuticals, Inc.
Editorial support for this poster was provided by Second City Science, LLC.

Additional Information

For more information on this study, visit www.zentalis.com or contact medicalaffairs@zentalis.com.

Abbreviations

1L, first line; 2L, second line; 5:2, 5 days on, 2 days off; BRCA, BReast CAncer gene; CA-125, cancer antigen 125; CBR, clinical benefit rate; CDK, cyclin-dependent kinase; CR, complete response; DL, dose level; DLT, dose limiting toxicity; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group Performance Status; EOT, end of treatment; FIGO, International Federation of Gynecology and Obstetrics; G1-S, GAP1-synthesis; G2-M, GAP2-mitosis; HGSOC, platinum-resistant high-grade serous ovarian cancer; IV, intravenous; OC, ovarian cancer; ORR, objective response rate; PARPi, poly (ADP-ribose) polymerase inhibitor; PD, progressive disease; PFS, progression-free survival; PK, pharmacokinetics; PO, oral; PR, partial response; QD, daily; SD, stable disease; VEGF, vascular endothelial growth factor.

PRESENTED AT: European Society of Gynecologic Oncology Annual Meeting, February 26-28, 2026, Copenhagen, Denmark