Poster #3902

WEE1 Inhibition as a Therapeutic Strategy in Triple-Negative Breast Cancer: Evaluating Single Agent and Combination Activity of Azenosertib in Preclinical Models

Catherine Lee, Alexandra Levy, Mona Abed, Heekyung Chung, Olivier Harismendy, Doris Kim

Zentalis Pharmaceuticals, Inc. San Diego, CA, USA

BACKGROUND

- WEE1 kinase maintains genomic integrity by regulating the G1-S and G2-M cell cycle checkpoints to prevent cells with damaged DNA from progressing to the next phase
- Azenosertib is a potent and selective oral WEE1 inhibitor currently in clinical development
- Treating tumor cells with azenosertib promotes premature cell cycle progression, leading to increased replication stress, accumulation of DNA damage, and ultimately mitotic catastrophe and cell death[1]
- Mechanisms such as *TP53* inactivation and Cyclin E1 overexpression often disrupt the G1-S checkpoint in tumors, increasing intrinsic replication stress and reliance on the G2-M checkpoint, thereby rendering tumor cells more vulnerable to WEE1 inhibition
- While clinically meaningful activity of azenosertib has been observed in Cyclin E1-positive platinum-resistant ovarian cancer (PROC)[2], other indications may have similarly high levels of replication stress and could benefit from WEE1 inhibition
- Triple-negative breast cancer (TNBC) represents 15%-20% of breast cancers diagnosed in the United States each year and is a particularly aggressive subtype with limited availability of targeted therapies for patients
- Although recent approvals of immune checkpoint inhibitors and antibody-drug conjugates (ADCs) have improved outcomes in some patients with TNBC, disease progression remains frequent, highlighting limitations such as ADC payload cross-resistance[3,4], and underscoring the need for new therapeutic strategies

Figure 1. Mechanism of action of azenosertib

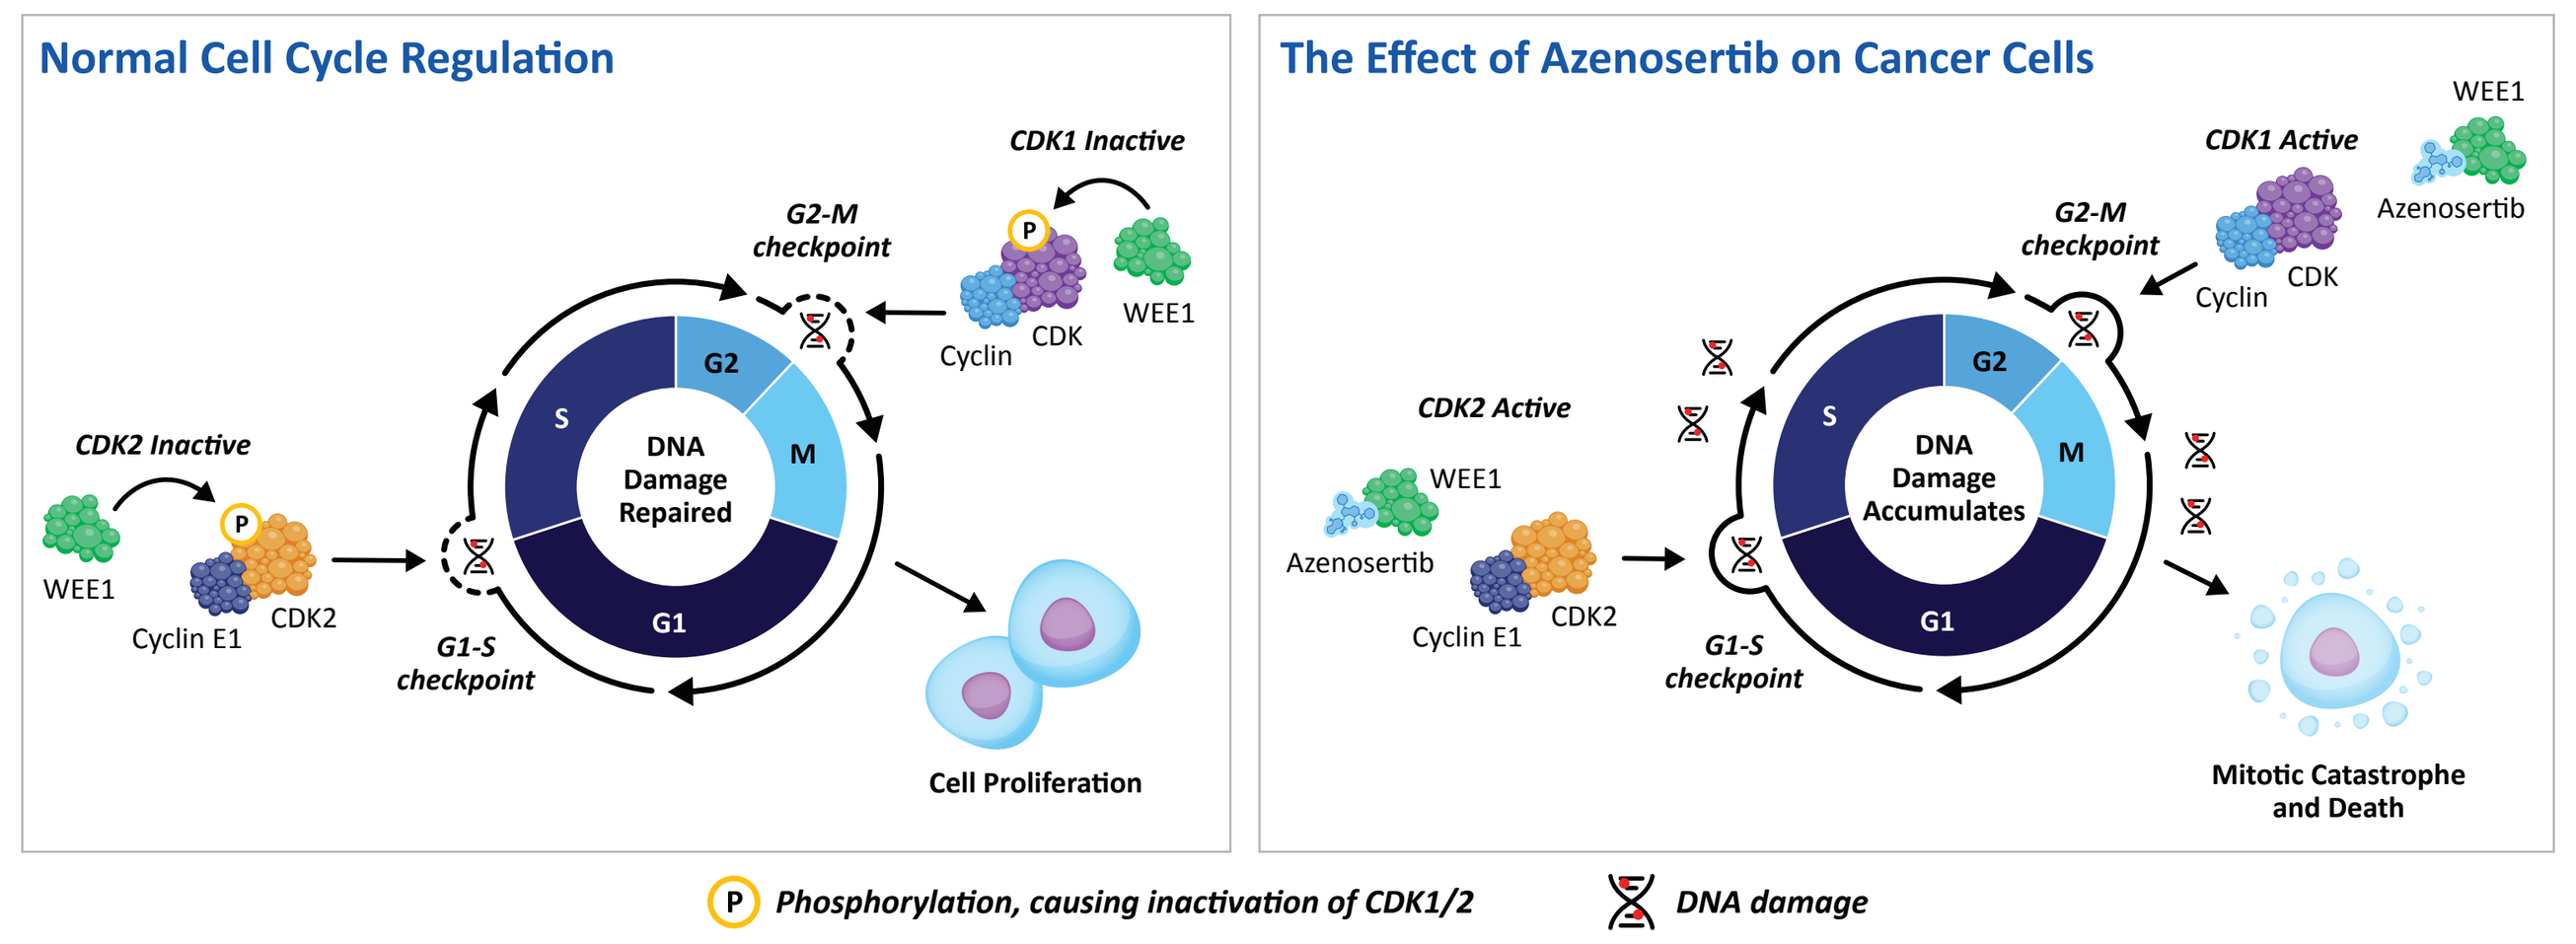

Normal Cell Cycle Regulation
CDK1 Inactive
G2-M checkpoint
WEE1
Cyclin
CDK
CDK2 Inactive
WEE1
Cyclin E1
CDK2
G1-S checkpoint
S
G2
M
G1
DNA Damage Repaired
Cell Proliferation
The Effect of Azenosertib on Cancer Cells
CDK1 Active
WEE1
Azenosertib
G2-M checkpoint
Cyclin
CDK
CDK2 Active
WEE1
Azenosertib
Cyclin E1
CDK2
G1-S checkpoint
DNA Damage Accumulates
Mitotic Catastrophe and Death
Phosphorylation, causing inactivation of CDK1/2
DNA damage

Figure 2. ADCs have the potential to reshape the treatment landscape of TNBC

Disease progression and cross-resistance to similar payloads are becoming relevant unmet needs in the clinic

1L
- Pembrolizumab + chemotherapy (if PD-L1+)
- Chemotherapy, often taxane/platinum (if PD-L1-)
- PARPi (if g*BRCA*+)
- **Ph3 ASCENT-04:** Sacituzumab govitecan + pembrolizumab (if PD-L1+)
- **Ph3 TROPION-Breast05:** Datopotamab deruxtecan + durvalumab (if PD-L1+)
- **Ph3 ASCENT-03:** Sacituzumab govitecan (if PD-L1-)
- **Ph3 TROPION-Breast02:** Datopotamab deruxtecan (if PD-L1-)

2L
- Sacituzumab govitecan
- Trastuzumab deruxtecan (if HER2-low)
- PARPi (if g*BRCA*+ and not used previously)
- Other biomarker-directed, targeted therapies

3L
- Chemotherapy
- Trastuzumab deruxtecan (if HER2-low and not used previously)
- PARPi (if g*BRCA*+ and not used previously)
- Other biomarker-directed, targeted therapies

- Topoisomerase 1 inhibitor (TOP1i) payload ADCs are currently approved for treatment of TNBC in the ≥2L setting and are rapidly advancing toward 1L use
- Sequential treatment with ADCs bearing the same payload class has demonstrated limited clinical benefit[3,4], highlighting the need for alternative post-ADC treatment strategies

CONCLUSIONS

- **TNBCs likely have high replication stress and are sensitive to WEE1 inhibition *in vitro* and *in vivo***
 - Most ~TNBC exhibited high Cyclin E1 protein expression and *TP53* mutation, suggesting dysregulated G1-S checkpoint control and high levels of replication stress in this patient population
 - Compared to non-TNBC breast cancer cell lines, TNBC cell lines expressed higher levels of Cyclin E1 and showed increased sensitivity to pharmacologic inhibition of WEE1 *in vitro*
 - Azenosertib demonstrated meaningful *in vivo* monotherapy activity (range, 42%-99% TGI) across a molecularly diverse panel of 12 TNBC xenograft models
 - In ADC-naïve models, combination treatment with azenosertib and TOP1i-payload ADCs (SG, Dato-DXd, or T-DXd) further enhanced antitumor efficacy, improving both depth and duration of response over the monotherapy treatments
- **Azenosertib combinations may be promising therapeutic strategies for TOP1i-ADC–resistant TNBC**
 - Azenosertib restored sensitivity to paclitaxel when combined in a PDX from a heavily pretreated TNBC patient (including prior paclitaxel and a TOP1i-ADC)
 - Azenosertib + EV combination led to a majority of complete responses in a TOP1i-ADC–resistant TNBC model
 - Azenosertib + EV combination substantially delayed relapse, while single-agent EV treatment led to relapse at least twice as quickly and more frequently in a TOP1i-ADC–resistant model
 - **Combination of azenosertib with EV has potential to improve the clinical efficacy observed with EV alone in patients with breast cancer[11] (NCT04225117), especially in patients with TNBC previously treated with SG**

PRESENTED AT: American Association for Cancer Research (AACR), April 17-22, 2026, San Diego, California, USA

METHODS

Analysis of pharmacogenomic and proteomic datasets

- Comparison of Cyclin E1 protein levels in serous ovarian cancer (SOC) and basal-like breast cancer was determined from TCGA pan-cancer analysis.[5] Although not all basal-like breast cancers are TNBC, they demonstrate substantial molecular and histological overlap[6] and are consequently enriched for TNBC. For simplicity, we refer to this population as ~TNBC
- Breast cancer cell lines were classified as TNBC or non-TNBC, based on molecular and histological features and literature sources[6-8]
- *In vitro* sensitivity of breast cancer cell lines to WEE1i was evaluated using a cell-based screen of the OncoPanel cancer cell lines (Eurofins). Cells were incubated with 9 serial dilutions of azenosertib for 5 days. Comparative screening data for adavosertib was obtained from the Genomics of Drug Sensitivity in Cancer (GDSC) database[9]
- Cyclin E1 protein levels in breast cancer cell lines were determined using proteomic data available from the Cancer Cell Line Encyclopedia (CCLE)[10]

***In vivo* animal studies**

- Monotherapy efficacy of azenosertib was evaluated in a panel of 12 TNBC CDX and PDX models
- Combinations of azenosertib with ADCs carrying TOP1i payloads (SG, Dato-DXd, and T-DXd were tested in two ADC-naïve models, MDA-MB-231 (CDX) and CTG-3103 (PDX)
- Combinations of azenosertib with chemotherapy (paclitaxel) and, separately, azenosertib with a microtubule inhibitor (MTI) payload ADC (EV) were tested in a TOP1i payload ADC-resistant PDX model of TNBC
- All tumor models were implanted subcutaneously in immunocompromised mice. Azenosertib was delivered orally, paclitaxel was delivered intraperitoneally, and all ADCs were delivered intravenously

Table 1. ADCs used in this study

ADC	Abbreviation	Target	Payload	Payload Class
Sacituzumab govitecan	SG	TROP2	SN38	Topoisomerase 1 inhibitor
Datopotamab deruxtecan	Dato-DXd	TROP2	DXd	Topoisomerase 1 inhibitor
Trastuzumab deruxtecan	T-DXd	HER2	DXd	Topoisomerase 1 inhibitor
Enfortumab vedotin	EV	Nectin-4	MMAE	Microtubule inhibitor

RESULTS

Figure 3. Basal-like breast cancers (~TNBC) exhibit molecular features consistent with impaired G1-S cell cycle checkpoint regulation

***CCNE1* amplification, Cyclin E1 expression, *TP53* mutation in TCGA pan-cancer dataset[5]**

Cancer Type	Total N	Prevalence *CCNE1* Amplified (%)	Prevalence Cyclin E1 Positive (%)	Prevalence *TP53* Mutated (%)
Serous ovarian cancer (SOC)	405	76/405 (19%)	253/405 (62%)	277/405 (68%)
Basal-like Breast cancer (~TNBC)	129	19/129 (15%)	123/129 (95%)	113/129 (88%)

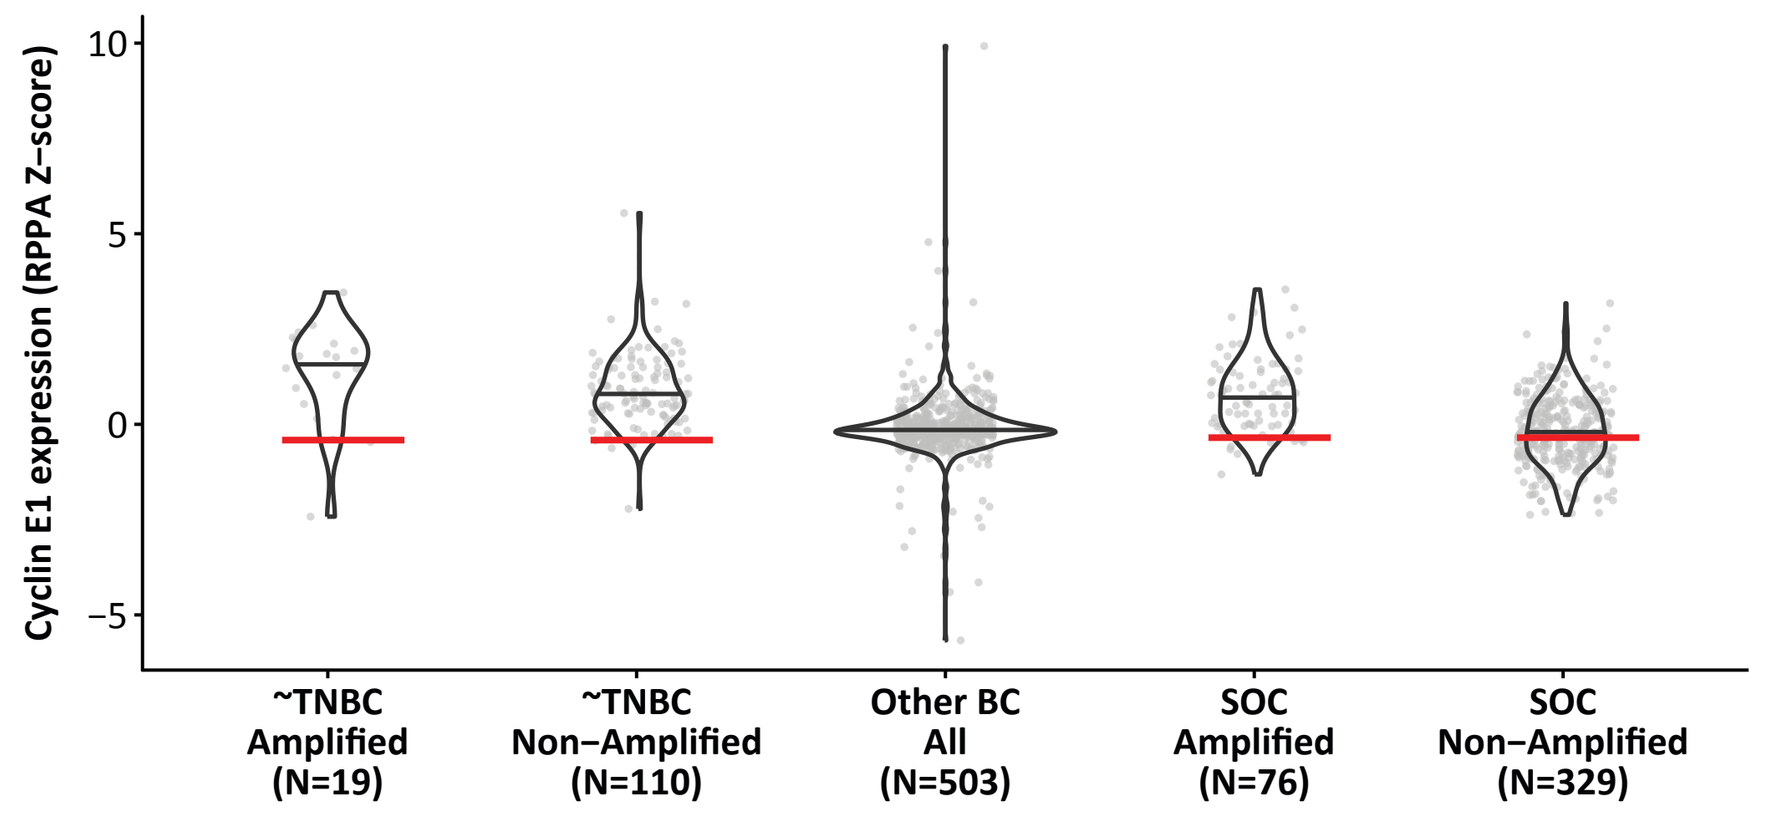

Cyclin E1 expression (RPPA Z-score)
10
5
0
−5
~TNBC Amplified (N=19)
~TNBC Non−Amplified (N=110)
Other BC All (N=503)
SOC Amplified (N=76)
SOC Non−Amplified (N=329)

- High level of Cyclin E1 expression: Positive Cyclin E1 expression observed in most *CCNE1* non-amplified tumors and in 95% and 62% of ~TNBC and SOC cases, respectively
- Frequent loss of *TP53* observed in 88% and 68% of ~TNBC and SOC cases, respectively
- Weakened G1-S cell cycle checkpoint may result in high replication stress and vulnerability to WEE1 inhibition

Distribution of Cyclin E1 protein expression levels (Z-score) from analysis of RPPA data in SOC and ~TNBC patients from TCGA pan-cancer analysis. *CCNE1* amplified and non-amplified are separated, and red segments denote the indication-specific expression level of the 10th percentile of *CCNE1* amplified sample ("TPA cutoff"). Expression levels above TPA cutoff are considered positive.

Figure 4. TNBC cell lines are more sensitive to WEE1 inhibition and exhibit higher Cyclin E1 protein expression compared with non-TNBC cell lines

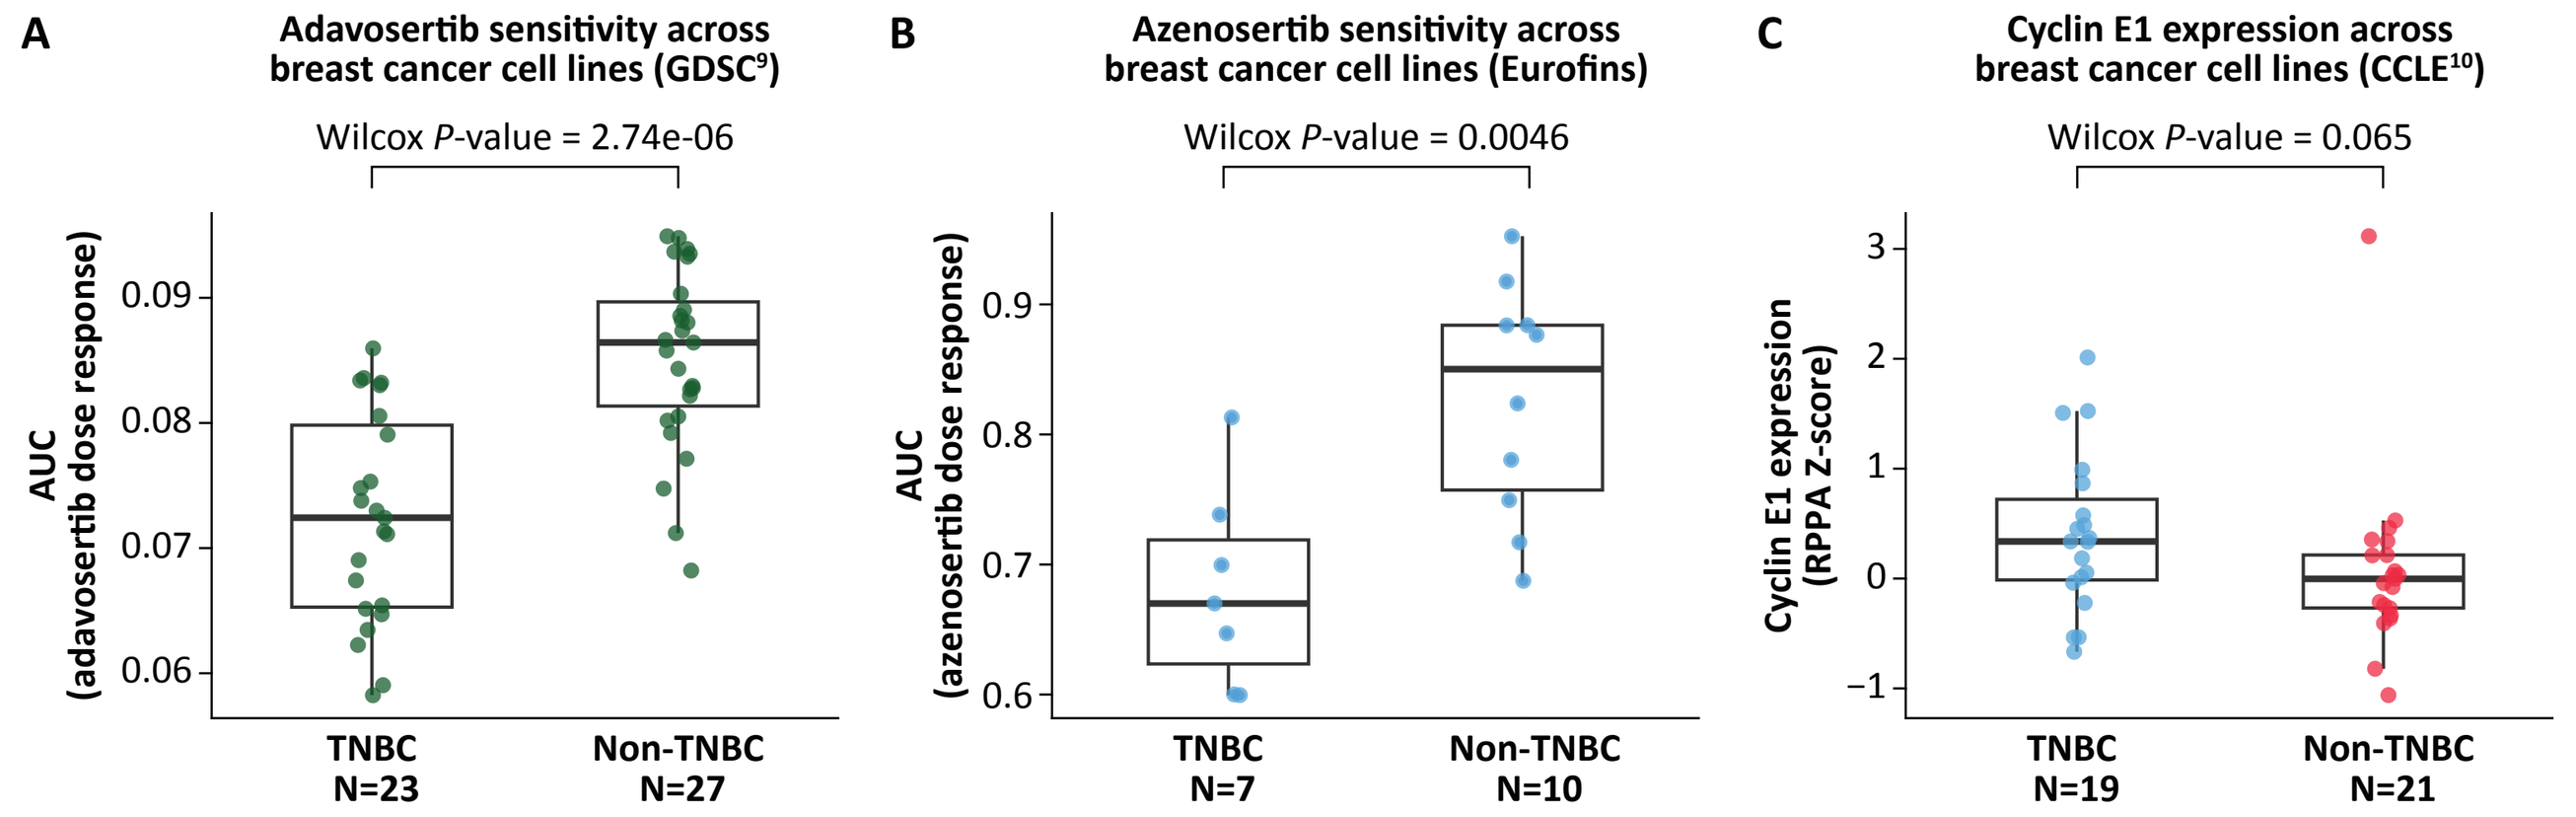

A
Adavosertib sensitivity across breast cancer cell lines (GDSC[9])
Wilcox P-value = 2.74e-06
AUC (adavosertib dose response)
TNBC N=23
Non-TNBC N=27
B
Azenosertib sensitivity across breast cancer cell lines (Eurofins)
Wilcox P-value = 0.0046
AUC (azenosertib dose response)
TNBC N=7
Non-TNBC N=10
C
Cyclin E1 expression across breast cancer cell lines (CCLE[10])
Wilcox P-value = 0.065
Cyclin E1 expression (RPPA Z-score)
TNBC N=19
Non-TNBC N=21

A. Distribution of area under the curve (AUC) values for breast cancer cell lines treated with adavosertib in the GDSC dataset, where lower AUC indicates increased drug sensitivity. **B.** Distribution of AUC values for breast cancer cell lines treated with azenosertib in the Eurofins dataset, where lower AUC indicates increased drug sensitivity. **C.** Distribution of Cyclin E1 protein expression levels (Z-score) from analysis of RPPA data in TNBC and non-TNBC breast cancer cell lines from the CCLE dataset.

Figure 5. Clinically relevant doses of azenosertib demonstrate monotherapy antitumor activity across a diverse panel of TNBC xenograft models *in vivo*

A

Model	Molecular Characteristics
SUM149PT	*TP53* mut; *BRCA1* mut (Δ11q)
HCC1937	*TP53* mut; *BRCA1* mut; *FAM35A* del; *RB1* mut
MDA-MB-468	*TP53* mut; *RB1* mut
HBCx-17	*TP53* mut; *BRCA2* mut; *AKT1* mut; *CDKN2A* mut; *KDM6A* del
MDA-MB-436	*BRCA1* mut; *RB1* mut
HCC1806	*TP53* mut
MDA-MB-231	*TP53* mut; *KRAS* G13D
HBCx-10	*TP53* mut; *BRCA2* mut; *RB1* del; *PTEN* del
HBCx-9	*TP53* mut
CTG-3103	*TP53* mut; *CCNE1* amp; *MYC* amp

B

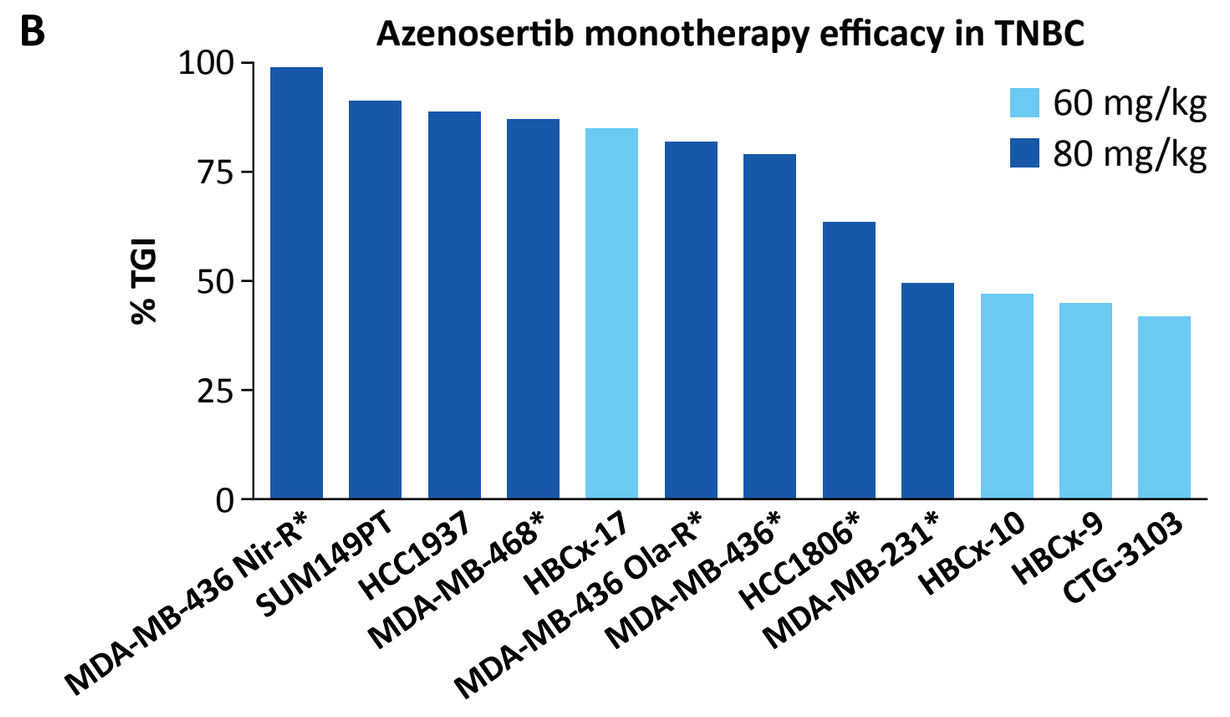

Azenosertib monotherapy efficacy in TNBC
60 mg/kg
80 mg/kg
% TGI
MDA-MB-436 Nir-R*
SUM149PT
HCC1937
MDA-MB-468*
HBCx-17
MDA-MB-436 Ola-R*
MDA-MB-436*
HCC1806*
MDA-MB-231*
HBCx-10
HBCx-9
CTG-3103

TNBC xenograft models harboring diverse mutations and copy number alterations were treated *in vivo* with 60 or 80 mg/kg of azenosertib. **A.** Key molecular characteristics of each model. **B.** Antitumor efficacy of azenosertib across models, expressed as percent tumor growth inhibition (TGI), calculated using the formula (1 - [Td - T0] / [Cd - C0]) × 100%. HBCx-17, HBCx-10, HBCx-9, and CTG-3103 are PDX models; all other models are CDX. Models labeled "Nir-R" and "Ola-R" have derived resistance to niraparib and olaparib, respectively. Models labeled with * were dosed once daily (QD) continuously; all other models were dosed QD using a 5:2 schedule.

Figure 6. Combinations of azenosertib with TOP1i-payload ADCs enhance tumor growth inhibition and duration of response in TNBC xenograft models

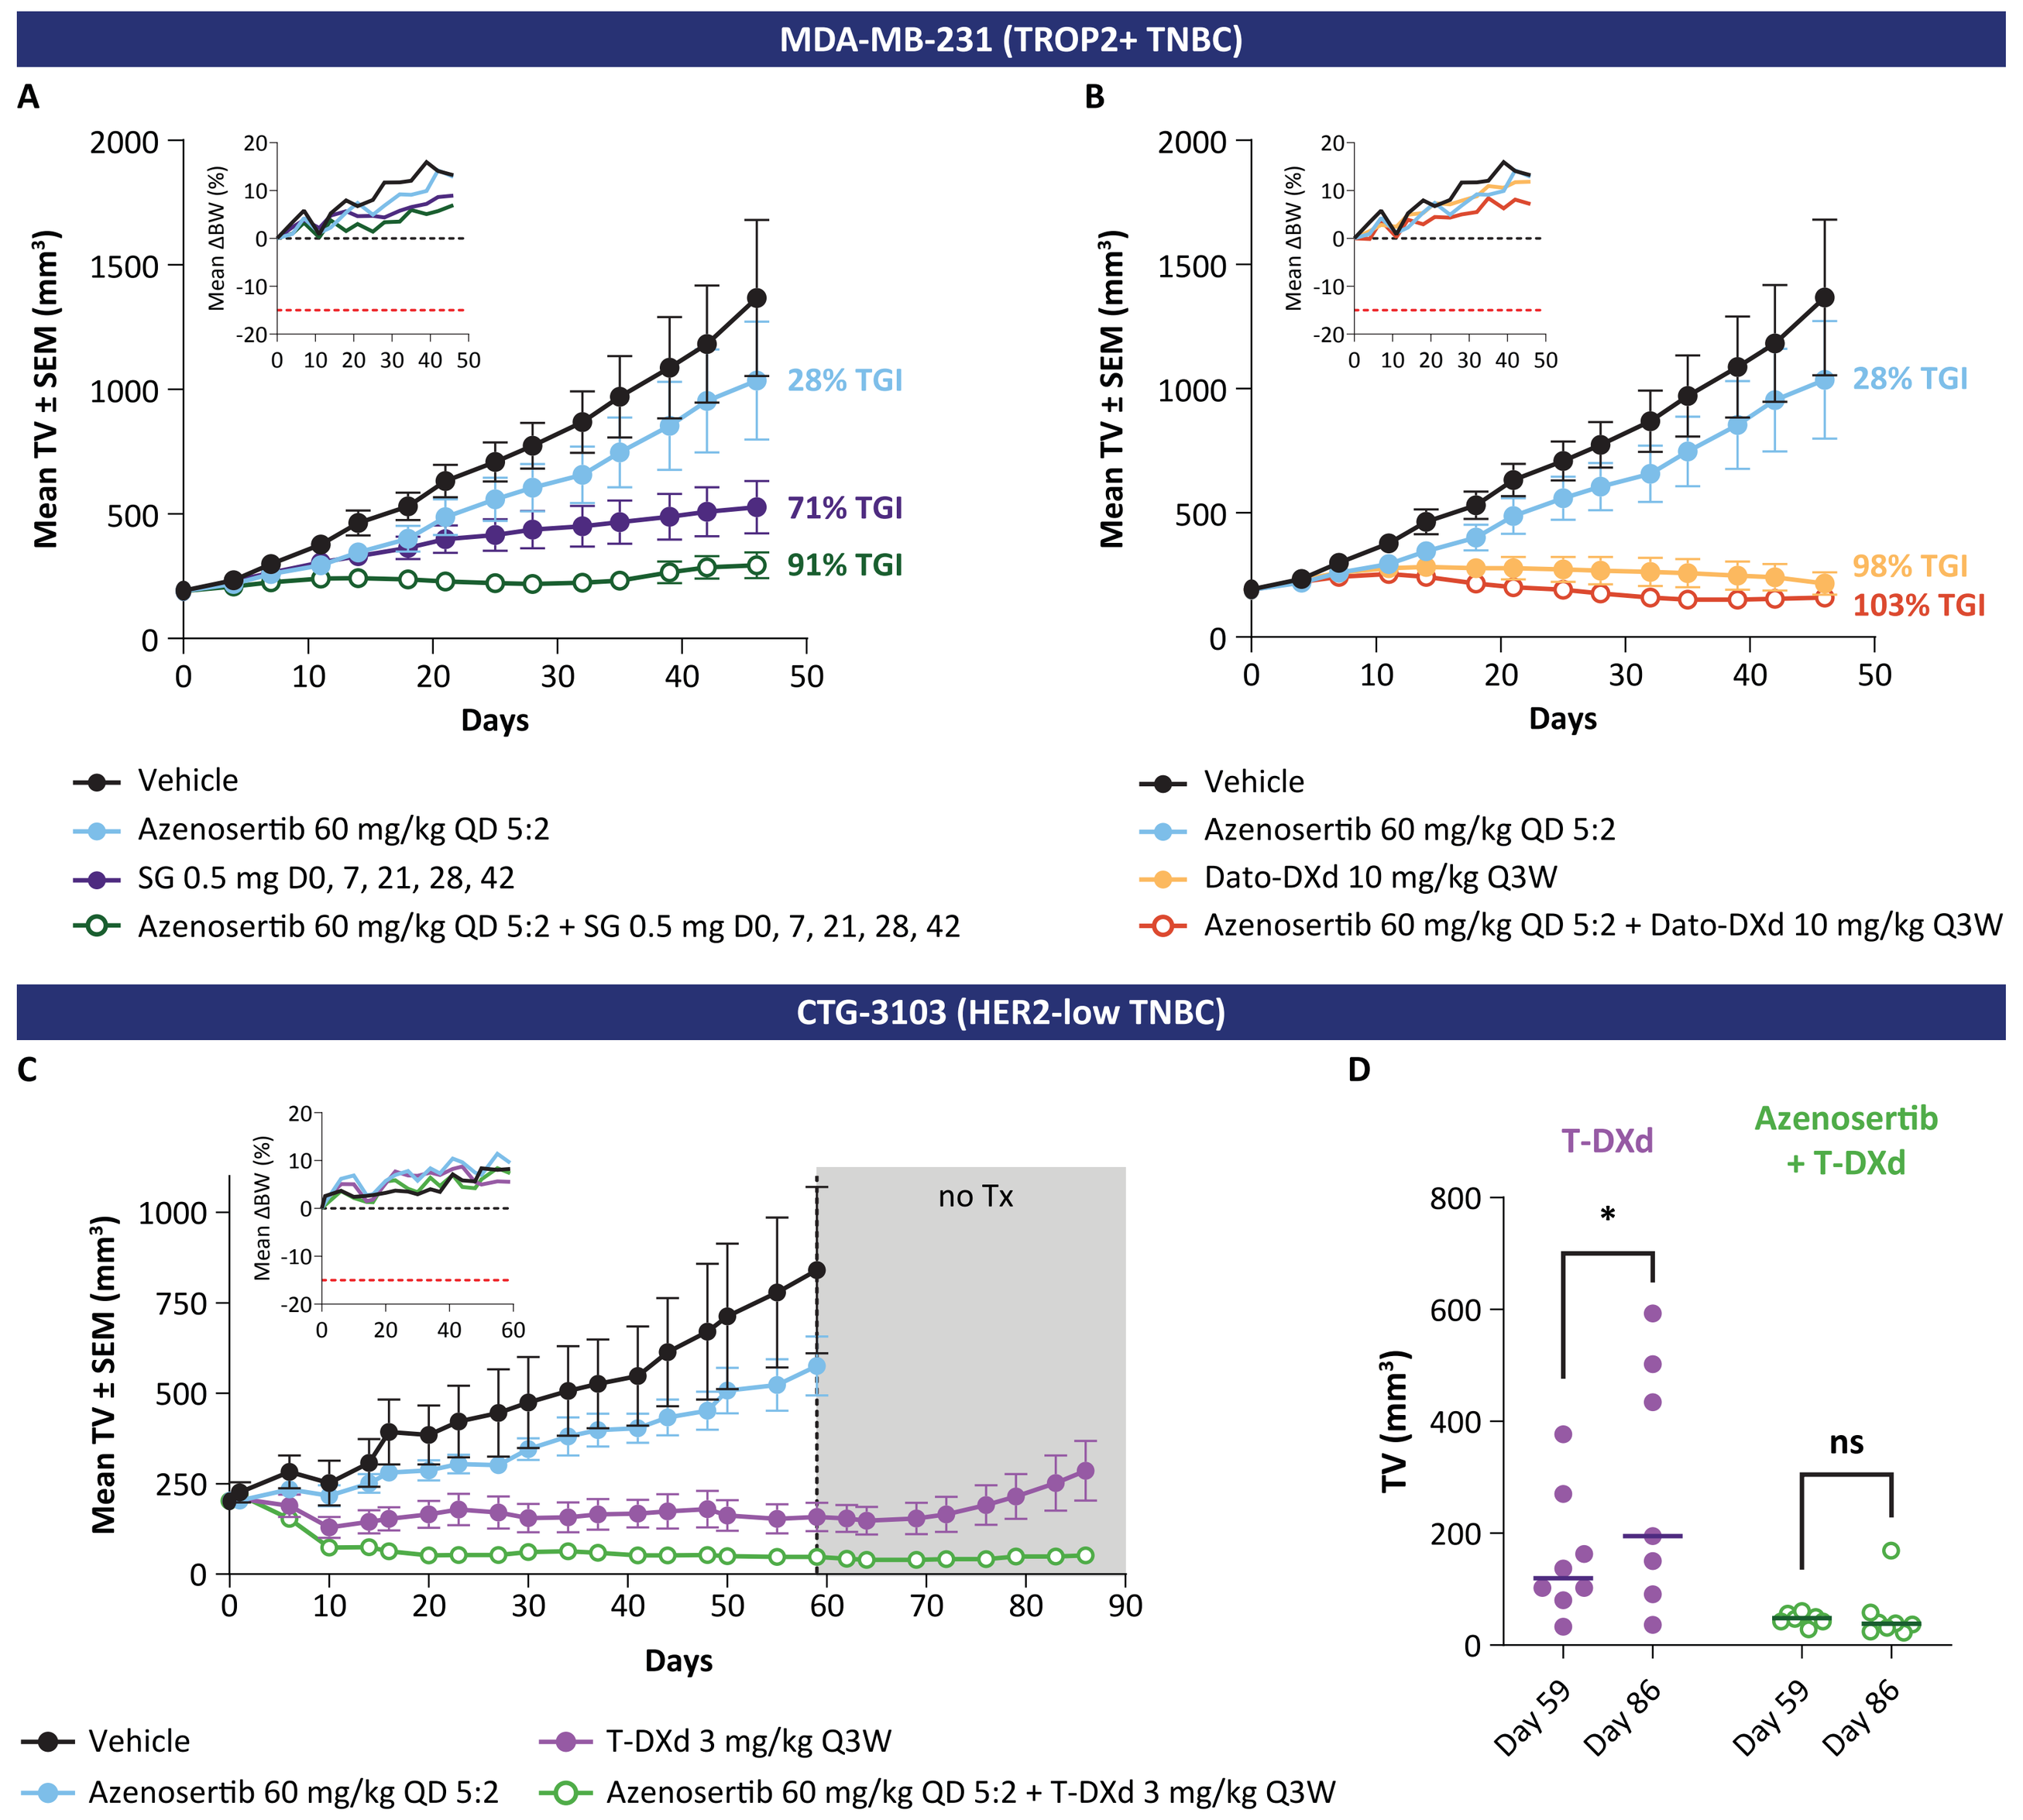

MDA-MB-231 (TROP2+ TNBC)
A
Mean TV ± SEM (mm³)
Mean ΔBW (%)
Days
28% TGI
71% TGI
91% TGI
Vehicle
Azenosertib 60 mg/kg QD 5:2
SG 0.5 mg D0, 7, 21, 28, 42
Azenosertib 60 mg/kg QD 5:2 + SG 0.5 mg D0, 7, 21, 28, 42
B
28% TGI
98% TGI
103% TGI
Vehicle
Azenosertib 60 mg/kg QD 5:2
Dato-DXd 10 mg/kg Q3W
Azenosertib 60 mg/kg QD 5:2 + Dato-DXd 10 mg/kg Q3W
CTG-3103 (HER2-low TNBC)
C
no Tx
Vehicle
Azenosertib 60 mg/kg QD 5:2
T-DXd 3 mg/kg Q3W
Azenosertib 60 mg/kg QD 5:2 + T-DXd 3 mg/kg Q3W
D
T-DXd
Azenosertib + T-DXd
TV (mm³)
*
ns
Day 59
Day 86

TNBC xenograft models treated with azenosertib in combination with ADCs carrying TOP1i payloads (n=8/group). **(A,B.)** Mean TV ± SEM of MDA-MB-231 CDX treated with the indicated doses of azenosertib and **(A)** SG or **(B)** Dato-DXd for 46 days. Antitumor efficacy is expressed as percent TGI, calculated using the formula (1 - [Td - T0] / [Cd - C0]) × 100%. Inset graphs depict mean percent change in body weight from day 0 (ΔBW), where the black dashed line indicates 0% change and the red dashed line indicates -15% change. **C.** Mean TV ± SEM of CTG-3103 PDX treated with the indicated doses of azenosertib and T-DXd. Treatment was discontinued on day 59, and tumor regrowth was monitored thereafter, indicated by the shaded gray box labeled "no Tx." Inset graph depicts mean percent change in body weight from day 0 (ΔBW), where the black dashed line indicates 0% change and the red dashed line indicates -15% change. **D.** Distribution of individual mouse tumor volumes on last day of treatment (day 59) compared to last day of monitoring for tumor regrowth (day 86). Significant tumor regrowth was detected in T-DXd group after treatment discontinuation, *P* value <0.016 by Wilcoxon matched-pairs test.

Azenosertib enhances the antitumor activity of ADCs in ADC-sensitive models

- Combination of azenosertib and SG (**Panel A**) or Dato-DXd (**Panel B**) was well-tolerated and increased TGI in MDA-MB-231 tumors
- Combination of azenosertib and T-DXd (**Panels C, D**) was well-tolerated and increased both TGI and durability of response in CTG-3103 tumors

Table 2. Patient treatment history of a TNBC PDX model with clinically derived resistance to SG

Line	Treatment	Treatment Duration and Response
1	Doxorubicin + cyclophosphamide	4 cycles
2	Paclitaxel + carboplatin	4 cycles, mastectomy with residual disease
3	Capecitabine	8 cycles, local recurrence
4	Multi-kinase inhibitor + nivolumab	1 cycle, disease progression
5	Bispecific anti-MUC1 x EGFR ADC	6 cycles, disease progression
6	Sacituzumab govitecan	2 cycles, disease progression

Figure 7. Combination of azenosertib with paclitaxel enhances TGI in a PDX model of SG-resistant TNBC

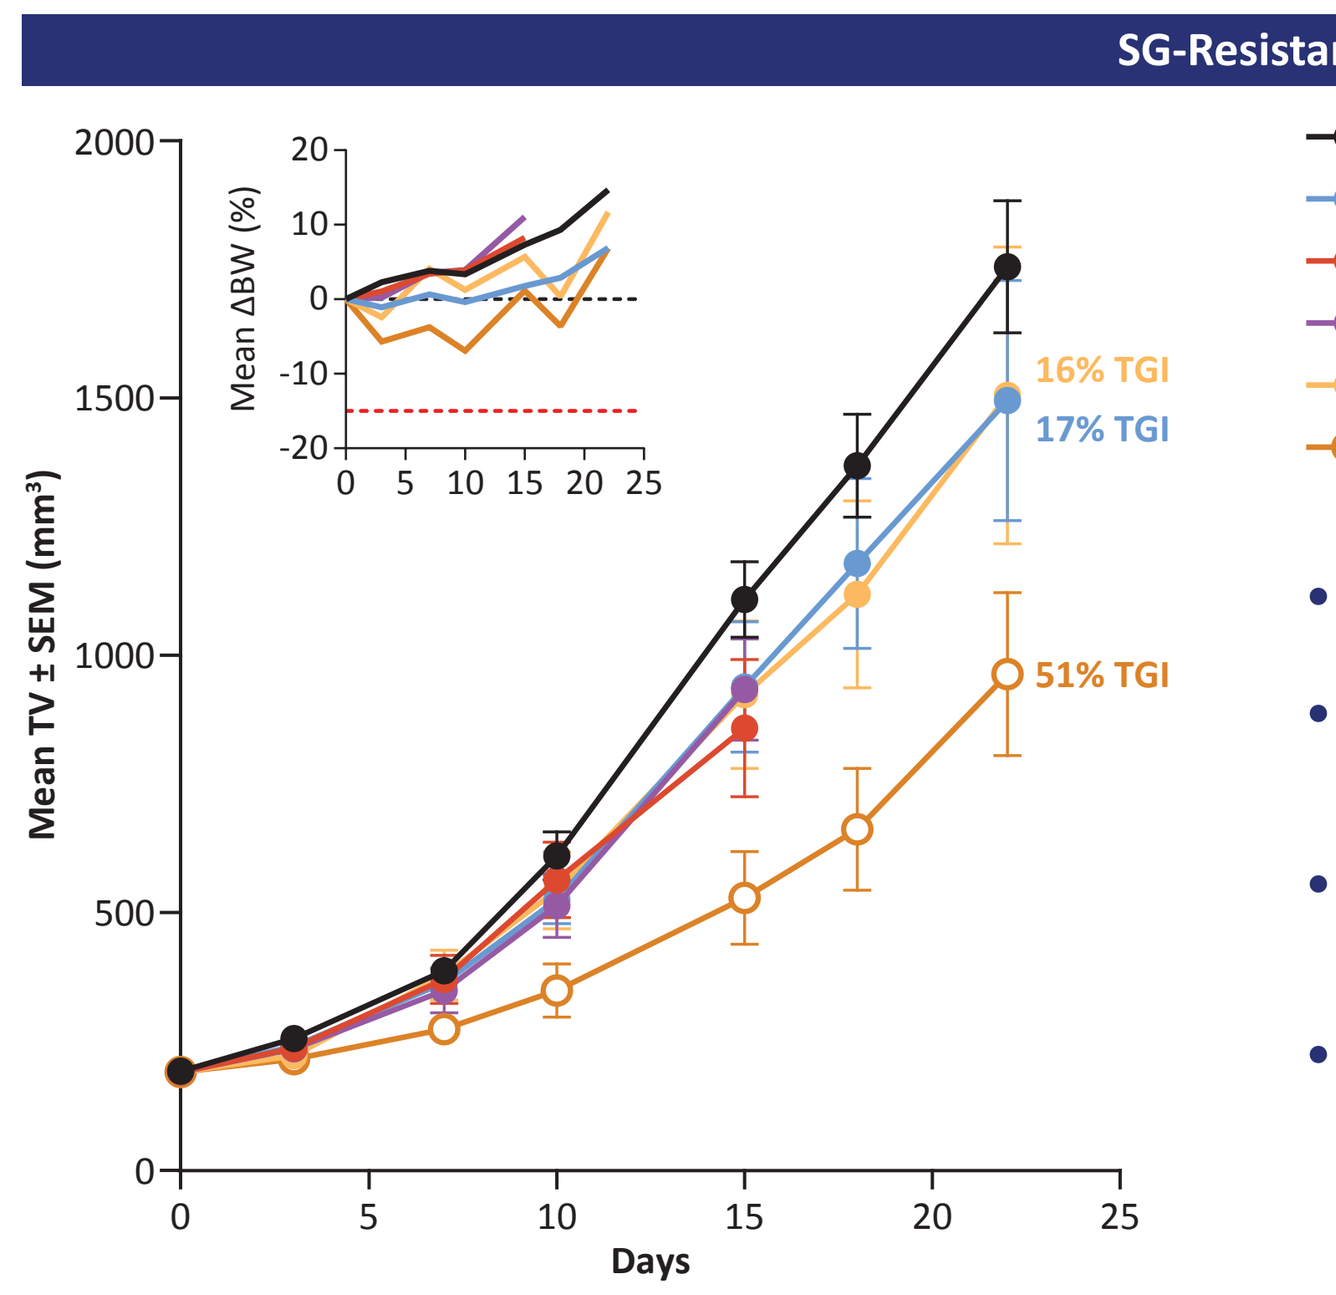

SG-Resistant TNBC
Mean TV ± SEM (mm³)
Mean ΔBW (%)
Days
16% TGI
17% TGI
51% TGI
Vehicle
Azenosertib 60 mg/kg QD 5:2
SG 3 mg/kg D1, 8
T-DXd 3 mg/kg D1
Paclitaxel 15 mg/kg QW
Azenosertib 60 mg/kg QD 5:2 + paclitaxel 15 mg/kg QW

- Control treatments with SG and T-DXd confirmed resistance to both SG and T-DXd, suggesting TOP1i payload cross-resistance
- In addition to prior SG, the patient received 4 cycles of paclitaxel treatment in 2L, which may explain the model's resistance to paclitaxel
- Although the model demonstrated resistance to monotherapy paclitaxel, combination treatment with azenosertib demonstrated potential to resensitize tumors to paclitaxel
- As TOP1i payload ADCs advance into 1L, combination of azenosertib and paclitaxel may be a promising therapeutic strategy post-ADC treatment

Mean TV ± SEM of an SG-resistant PDX model treated with azenosertib +/- paclitaxel (n=8/group). Antitumor efficacy is expressed as percent TGI, calculated using the formula (1 - [Td - T0] / [Cd - C0]) × 100%. Inset graph depicts mean percent change in body weight from day 0 (ΔBW), where the black dashed line indicates 0% change and the red dashed line indicates -15% change.

Figure 8. Combination of azenosertib with an MTI-payload ADC (EV) induces tumor regressions and complete responses in a TNBC PDX model resistant to a TOP1i-payload ADC (SG)

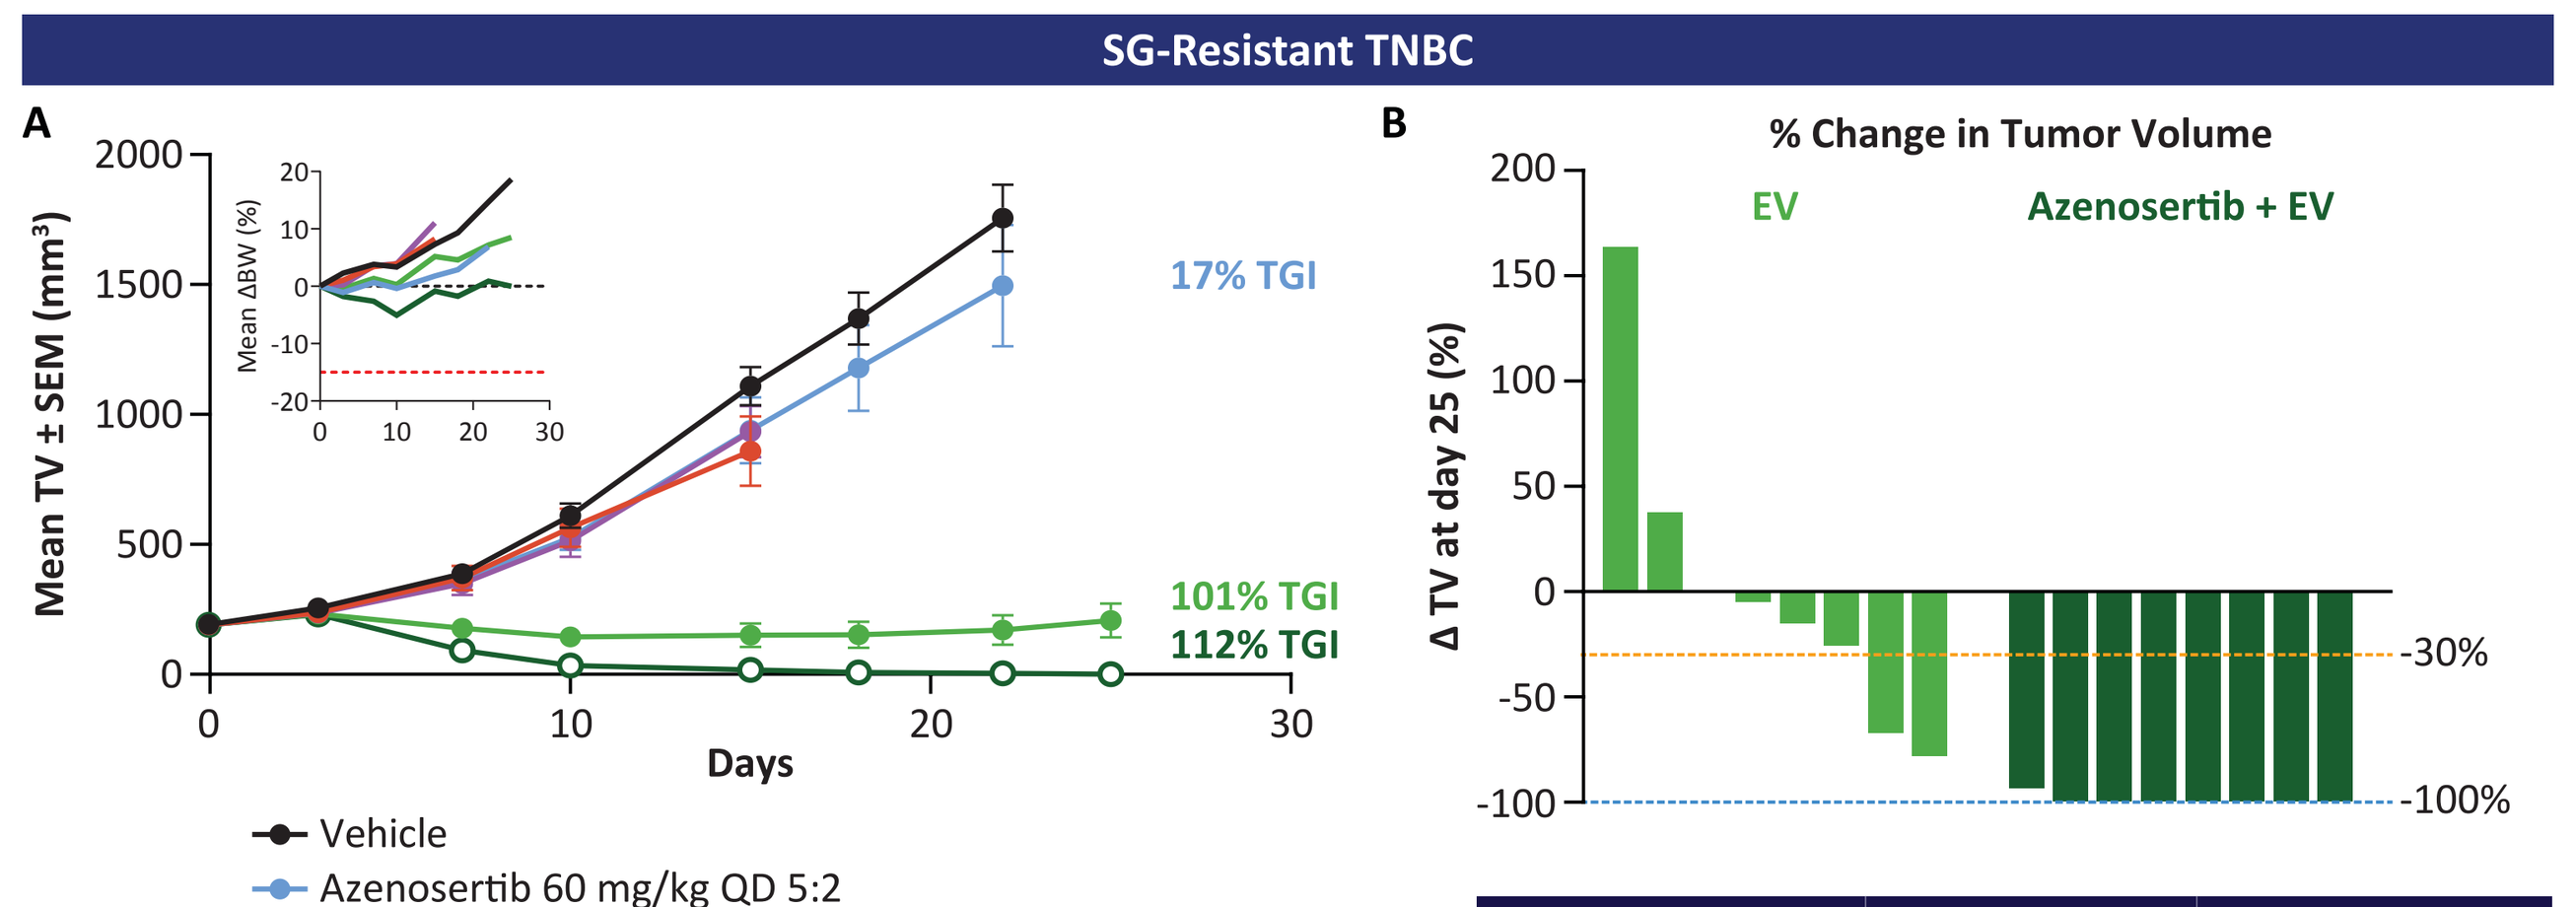

SG-Resistant TNBC
A
Mean TV ± SEM (mm³)
Mean ΔBW (%)
Days
17% TGI
101% TGI
112% TGI
Vehicle
Azenosertib 60 mg/kg QD 5:2
SG 3 mg/kg D1, 8
T-DXd 3 mg/kg D1
EV 3 mg/kg D1, 8, 15 (28d cycle)
Azenosertib 60 mg/kg QD 5:2 + EV 3 mg/kg D1, 8, 15
B
% Change in Tumor Volume
EV
Azenosertib + EV
Δ TV at day 25 (%)
-30%
-100%

Treatment Group	PR at Day 25	CR at Day 25
EV	2/8 (25%)	0/8 (0%)
Azenosertib + EV	1/8 (12.5%)	7/8 (87.5%)

A. Mean TV ± SEM of an SG-resistant PDX model treated with azenosertib +/- EV (n=8/group). Antitumor efficacy is expressed as percent TGI relative to the vehicle group on day 22, calculated using the formula (1 - [Td - T0] / [Cd - C0]) × 100%. Inset graph depicts mean percent change in body weight from day 0 (ΔBW), where the black dashed line indicates 0% change and the red dashed line indicates -15% change. **B.** Waterfall plot showing percent change in TV (ΔTV) for individual mice on the last day of treatment (day 25), calculated using the formula ([TVd – TV0] / TV0) × 100%. Orange dashed line denotes a change of -30%, corresponding to the threshold for partial response (PR). Blue dashed line denotes a change of -100%, corresponding to complete response (CR). Table summarizes the frequency of PRs and CRs observed on day 25; any PRs or CRs occurring before or after day 25 are not reflected here but can be seen in Fig 9B.

Figure 9. Combination of azenosertib with EV drives durable tumor responses and extends PFS in a PDX model of SG-resistant TNBC

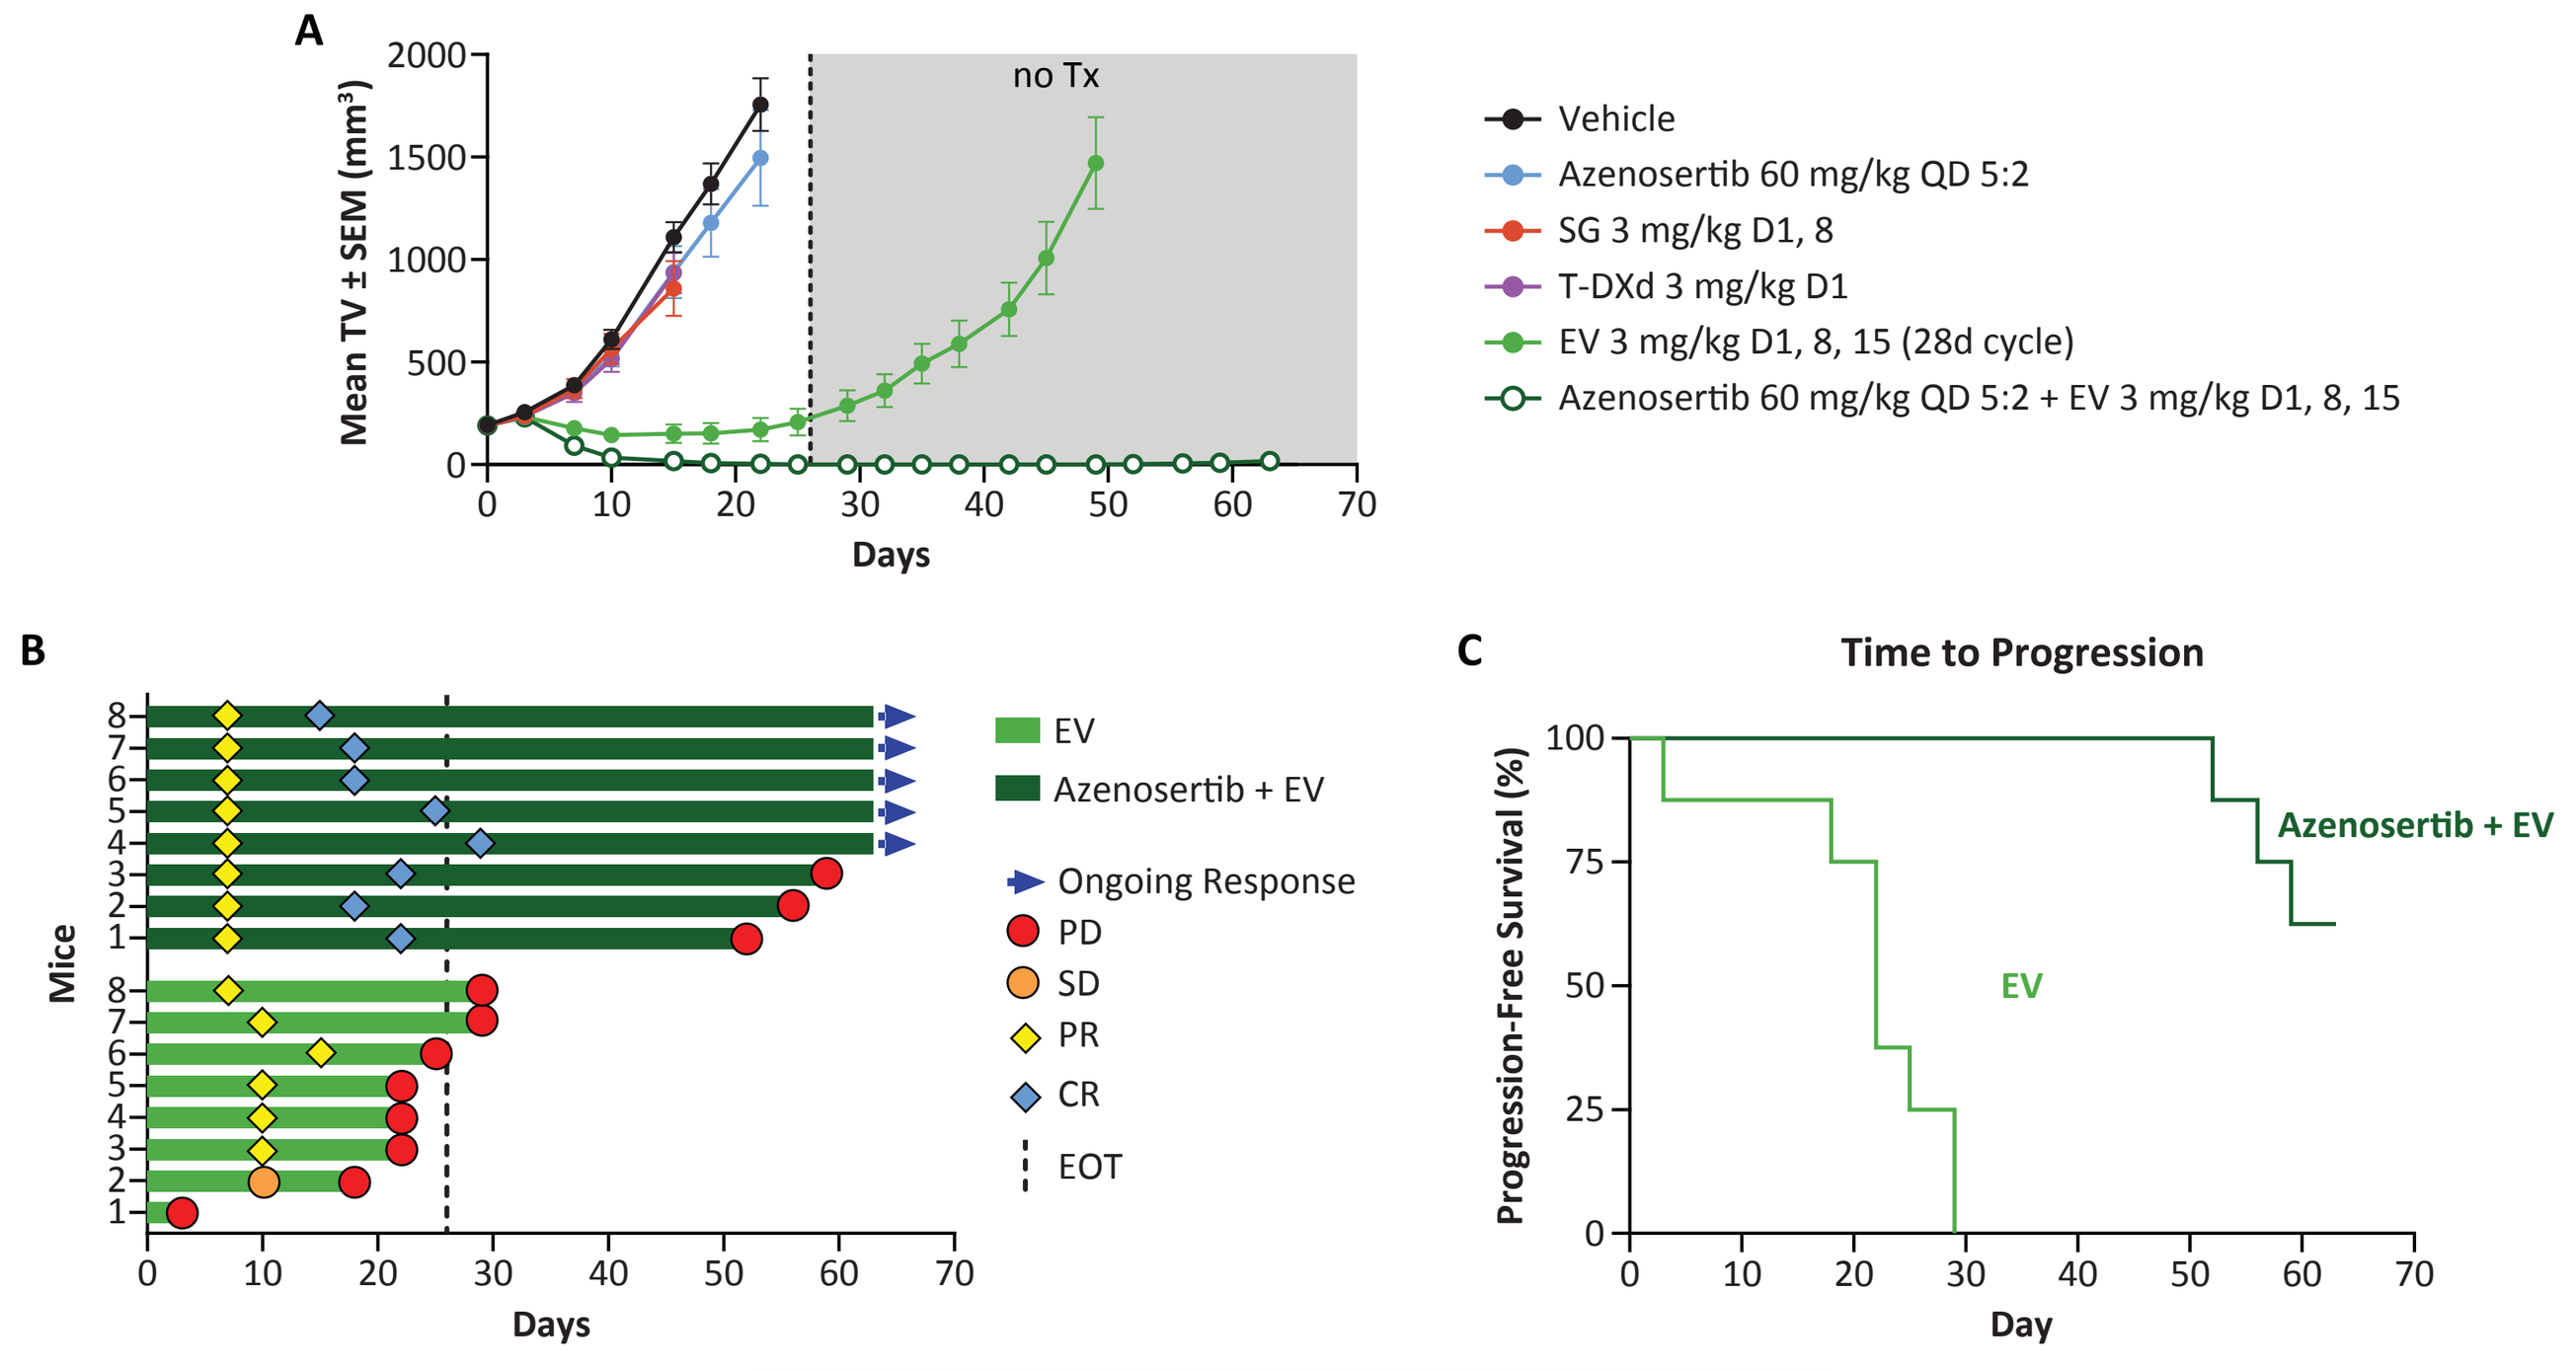

A
no Tx
Mean TV ± SEM (mm³)
Days
Vehicle
Azenosertib 60 mg/kg QD 5:2
SG 3 mg/kg D1, 8
T-DXd 3 mg/kg D1
EV 3 mg/kg D1, 8, 15 (28d cycle)
Azenosertib 60 mg/kg QD 5:2 + EV 3 mg/kg D1, 8, 15
B
Mice
Days
EV
Azenosertib + EV
Ongoing Response
PD
SD
PR
CR
EOT
C
Time to Progression
Progression-Free Survival (%)
Day
Azenosertib + EV
EV

Treatment Group	Median DoR	Median PFS
EV	12 days	22 days
Azenosertib + EV	>56 days	Not reached

A. Mean TV ± SEM of an SG-resistant PDX model treated with azenosertib +/- EV (n=8/group). Treatment was discontinued after day 25, and tumor regrowth was monitored thereafter, indicated by the shaded gray box labeled "no Tx." Observation of tumor regrowth in the azenosertib + EV group occurred in only 3 mice on days 52, 56, and 59, and mean TV on day 63 remained small at 17mm³. **B.** Swimmer plot depicting treatment response for individual mice. Dashed line indicates end of treatment (EOT) on day 25. **C.** Kaplan-Meier plot of PFS, defined as time from treatment initiation to documented disease progression, where progression is defined as >20% increase from the smallest recorded tumor volume: *P* value<0.0001 by log-rank (Mantel-Cox) test, HR: 0.14, 95% CI: 0.036-0.52. The table summarizes median duration of response (DoR) and median PFS. DoR was defined as the time from first PR to PD. Median DoR for combination group includes 5 ongoing responders at final measurement.

Figure 10. Sequential treatment of SG- or T-DXd-progressed tumors with azenosertib + EV provides robust antitumor activity compared to monotherapy EV

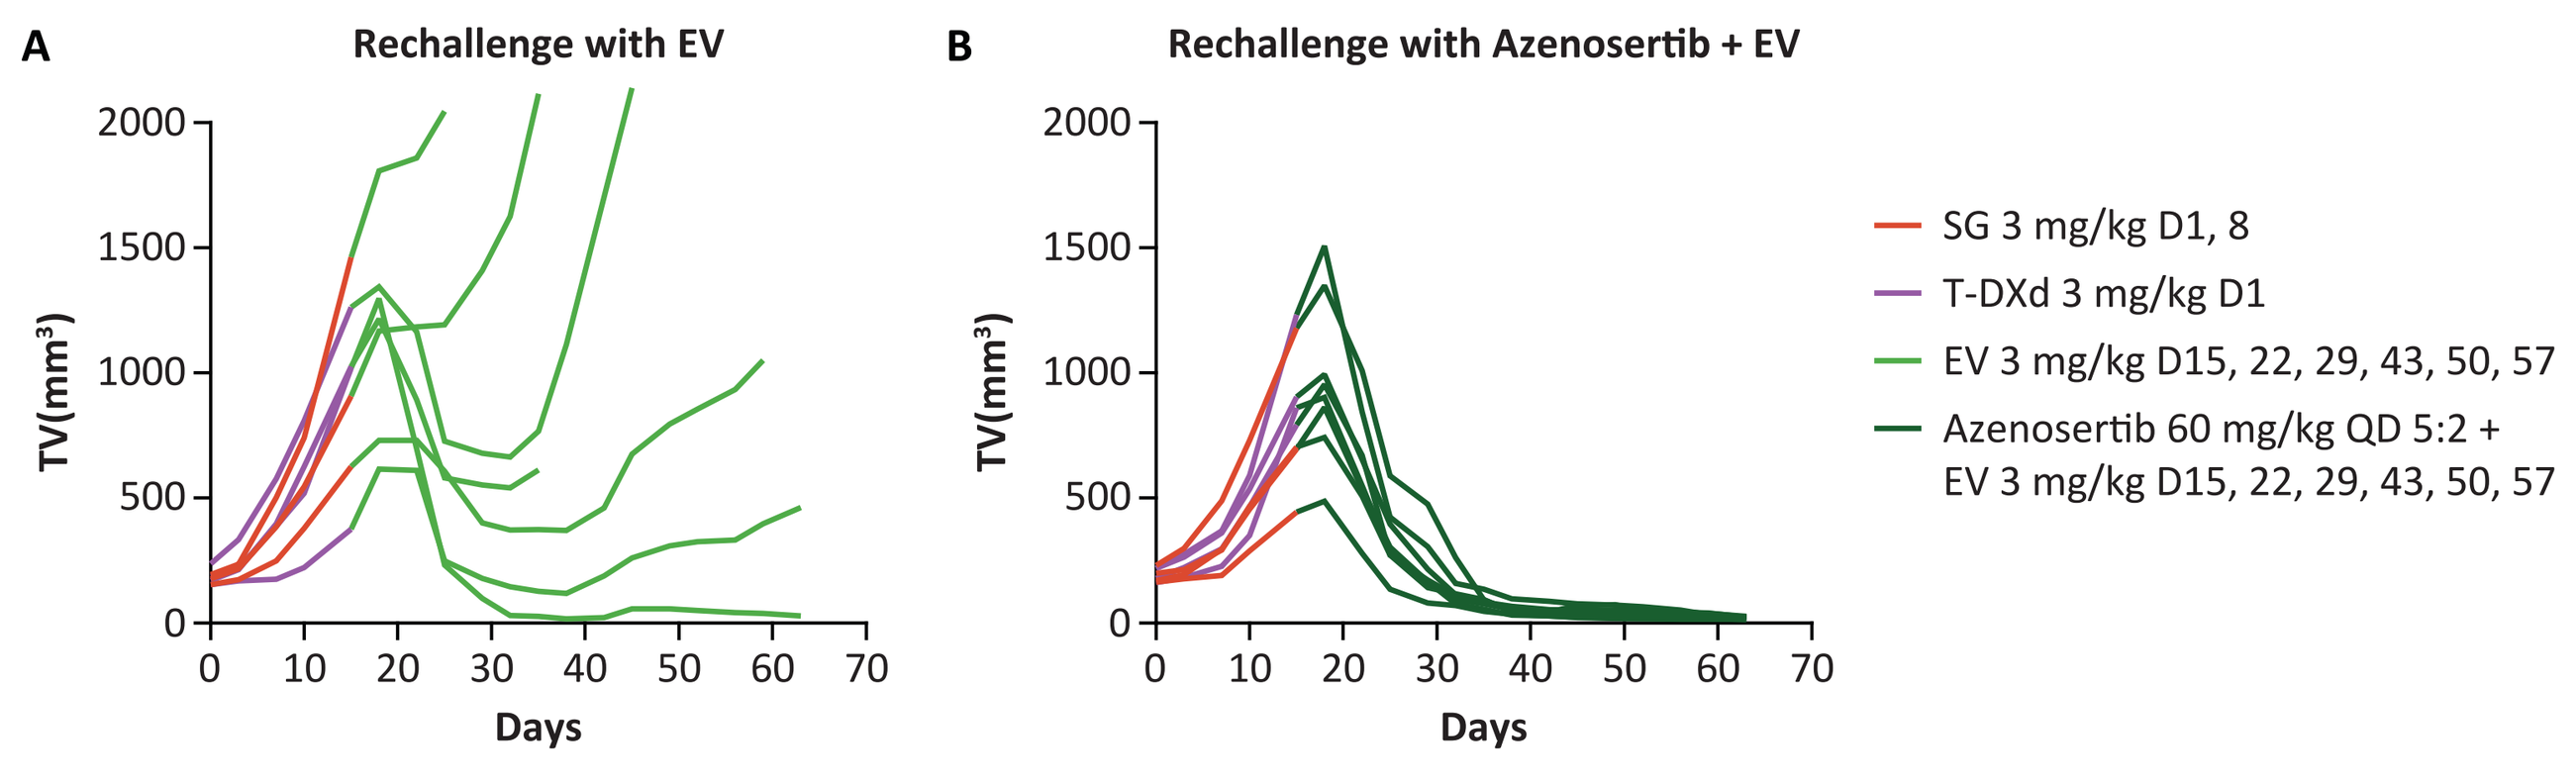

A
Rechallenge with EV
B
Rechallenge with Azenosertib + EV
TV(mm³)
Days
SG 3 mg/kg D1, 8
T-DXd 3 mg/kg D1
EV 3 mg/kg D15, 22, 29, 43, 50, 57
Azenosertib 60 mg/kg QD 5:2 + EV 3 mg/kg D15, 22, 29, 43, 50, 57

Spider plots displaying TVs of individual mice bearing SG-resistant PDX tumors that were first treated with SG or T-DXd for 14 days, rerandomized into new groups to minimize the impact of prior treatment, then rechallenged on day 15 with **(A)** monotherapy EV (n=7), or **(B)** combination of azenosertib + EV (n=8). Average TV on day 15 at the start of rechallenge was 850-950mm³. Treatment and tumor measurements continued until day 63.

References

1. Kim D, et al. *NPJ Precis Oncol*. 2025;9(1):3.
2. Simpkins F, et al. Presented at the SGO Annual Meeting 2025. Abstract 814654.
3. Tarantino P, et al. *J Natl Cancer Inst*. 2025;117(11):2327-2335.
4. Huppert LA, et al. *NPJ Breast Cancer*. 2025;11(1):34.
5. Akbani R, et al. *Nat Commun*. 2014;5:3887.
6. Dai X, et al. *J Cancer.* 2017;8(16):3131-3141.
7. Chavez KJ, et al. *Breast Dis*. 2010;32(1-2):35-48.
8. American Type Culture Collection 2022. Triple negative breast cancer panels. 2022. Accessed March 6, 2026. https://www.atcc.org/-/media/product-assets/documents/panels/cell-biology/triple-negative-breast-cancer-panels.pdf?rev=67ddbf2f83dd415d88f367b4b3ab9fc3 .
9. Garnett MJ, et al. *Nature*. 2012;483(7391):570-575.
10. Chen MM, et al. *JCO Clin Cancer Inform*. 2030;4:357-366.
11. Giordano A, et al. *J Clin Oncol*. 2024;42:16_suppl:1005-1005.

Acknowledgments

This study was sponsored by Zentalis Pharmaceuticals, Inc. Animal studies were conducted at Pharmaron (Ningbo/Beijing), Champions Oncology, and Crown Bioscience (Beijing).
Editorial support for this poster was provided by Second City Science, LLC.

Additional Information

For more information on this study, visit www.zentalis.com or contact publications@zentalis.com.

Abbreviations

1L, first line; 2L, second line; 3L, third line; 5:2, 5 days on, 2 days off; ADC, antibody-drug conjugate; Amp, amplified; AUC, area under the curve; BC, breast cancer; ΔBW, change in body weight; BRCA, breast cancer gene; CCLE, Cancer Cell Line Encyclopedia; CCNE1, Cyclin E1; CDK, cyclin-dependent kinase; CDX, cell line-derived xenograft; CI, confidence interval; CR, complete response; D, day; Dato-DXd, datopotamab deruxtecan; del, deletion; DoR, duration of response; EGFR, epidermal growth factor receptor; EOT, end of treatment; EV, enfortumab vedotin; gBRCA, germline BRCA; G1-S, GAP1-Synthesis; G2-M, GAP2-Mitosis; GDSC, Genomics of Drug Sensitivity in Cancer; HBCx, Human breast cancer xenograft; HER2, human epidermal growth factor receptor 2; HR, hormone receptor; KRAS, Kirsten rat sarcoma viral oncogene homolog; MMAE, monomethyl auristatin E; MTI, microtubule inhibitor; mut, mutation; Nectin-4, nectin cell adhesion molecule 4; ns, not significant; PARPi, poly (ADP-ribose) polymerase inhibitor; PD, progressive disease; PD-L1, programmed death-ligand 1; PDX, patient-derived xenograft; PFS, progression-free survival; Ph, phase; PR, partial response; QD, once daily; QW, once weekly; Q3W, once every 3 weeks; RB1, retinoblastoma 1; RPPA, reverse phase protein array; SD, stable disease; SEM, standard error of the mean; SG, sacituzumab govitecan; SOC, serous ovarian cancer; TCGA, The Cancer Genome Atlas; T-DXd, trastuzumab deruxtecan; TGI, tumor growth inhibition; TNBC, triple-negative breast cancer; TOP1i, topoisomerase 1 inhibitor; TP53, tumor protein 53; TPA, tenth percentile of amplification; TROP2, trophoblast cell surface antigen 2; ΔTV, change in tumor volume; TV, tumor volume; Tx, treatment.